Results from the Phase 1 Part of the PYNNACLE Study Assessing Rezatapopt in Patients with Advanced Solid Tumors Harboring a *TP53* Y220C Mutation

Amy Body,[1] Peter Grimison,[2] Michael Millward,[3] Shwagi Sukumaran,[4] Geoffrey I. Shapiro,[5] Aparna R. Parikh,[6] Melissa L. Johnson,[7] Anthony W. Tolcher,[8] John A. Thompson,[9] Anthony B. El-Khoueiry,[10] Andrae L. Vandross,[11] Shivaani Kummar,[12] Dale R. Shepard,[13] Kim LeDuke,[14] Marc Fellous,[11] Alison M. Schram,[15] Ecaterina E. Dumbrava[16]

[1]Monash Medical Centre, Clayton, VIC, Australia; [2]Chris O'Brien Lifehouse Hospital, Camperdown, NSW, Australia; [3]Linear Clinical Research, Nedlands, WA, Australia; [4]Flinders Medical Centre, Bedford Park, SA, Australia; [5]Dana Farber Cancer Institute, Boston, MA, USA; [6]Massachusetts General Hospital, Boston, MA, USA; [7]Sarah Cannon Research Institute, Nashville, TN, USA; [8]NEXT Oncology, San Antonio, TX, USA; [9]Fred Hutchinson Cancer Center, Seattle, WA, USA; [10]USC Norris Comprehensive Cancer Center, Los Angeles, CA, USA; [11]NEXT Oncology, Austin, TX, USA; [12]OHSU Knight Cancer Institute, Portland, OR, USA; [13]Cleveland Clinic, Cleveland, OH, USA; [14]PMV Pharmaceuticals, Inc., Princeton, NJ, USA; [15]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [16]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

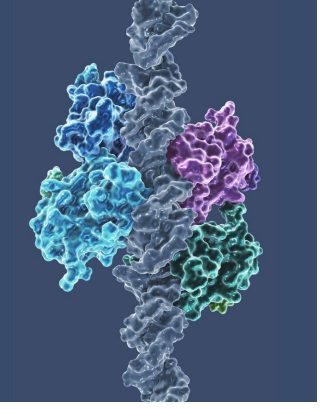

BACKGROUND

- *TP53* is a tumor suppressor gene encoding the p53 protein[1]
 - *TP53* mutations resulting in p53 inactivation are a key step in oncogenesis[1]
 - *TP53* mutations are found in ~59% of all solid tumors, including ~76% of ovarian cancer cases and ~51% of breast cancer cases[2–4]
 - They are associated with aggressive and invasive tumor types such as high-grade serous ovarian cancer (occurring in ~96% of all cases) and triple-negative breast cancers (occurring in 70–80% of cases), which have poorer outcomes[2,5]
- *TP53* Y220C is a missense mutation that occurs in ~1% of all solid tumors, including ~2.9% of ovarian cancers and ~1% of breast cancers[2,6]
 - The tyrosine to cysteine amino acid substitution destabilizes the p53 protein, leading to its inactivation[6]
- Reactivation of the wild-type p53 protein in p53-mutated tumors is an attractive therapeutic approach
- Rezatapopt (also known as PC14586) is an investigational, first-in-class p53 reactivator that selectively binds to the mutated p53 Y220C protein and stabilizes it in the wild-type conformation, thereby restoring p53 activity[6-8]
- PYNNACLE (NCT04585750) is a Phase 1/2 clinical trial of rezatapopt in patients with advanced solid tumors harboring a *TP53* Y220C mutation[9,10]

OBJECTIVE

- Here, we present results from the Phase 1 portion of PYNNACLE across the overall population and in subgroups of patients with ovarian and breast cancers

METHODS

- Patients eligible for the PYNNACLE Phase 1 study were ≥12 years of age with locally advanced or metastatic solid tumors harboring a *TP53* Y220C mutation
- Rezatapopt was administered in increasing oral doses for 21-day continuous cycles to determine the efficacious dose range (1150 mg once daily [QD] to 1500 mg twice daily [BID])
- Safety and preliminary efficacy were assessed by the investigator using Common Terminology Criteria for Adverse Events v5.0 and Response Evaluation Criteria in Solid Tumors v1.1, respectively
- The efficacy-evaluable population included patients who had a *TP53* Y220C mutation and were *KRAS* wild type (did not harbor a *KRAS* single-nucleotide variant) with measurable disease at baseline and ≥1 post-baseline tumor assessment receiving rezatapopt in the efficacious dose range
- Data cut-off: 5 September 2023

RESULTS

- As of 5 September 2023, 67 patients with various solid tumors received rezatapopt in the efficacious dose range (1150 mg QD to 1500 mg BID, **Table 1**)

Table 1. Patient demographics and disease characteristics

	Patients treated in the efficacious dose range of 1150 mg QD to 1500 mg BID		
	All patients (n=67)	**Ovarian cancer subgroup (n=22)**	**Breast cancer subgroup (n=9)**
Age, years			
Median (min-max)	63 (32–84)	66 (49-81)	53 (32-65)
Sex, n (%)			
Female	41 (61)	22 (100)	9 (100)
Male	26 (39)	0	0
Race, n (%)			
White	51 (76)	15 (68)	6 (67)
Asian	5 (7)	3 (14)	1 (11)
Black or African American	6 (9)	2 (9)	1 (11)
Other	1 (1)	0	0
Not reported/unknown	4 (6)	2 (9)	1 (11)
ECOG PS, n (%)			
0	22 (33)	6 (27)	3 (33)
1	45 (67)	16 (73)	6 (67)
Prior systemic therapies			
1	6 (9)	1 (5)	0
2	19 (28)	4 (18)	2 (22)
≥3	37 (55)	14 (64)	7 (78)
Not reported	5 (6)	3 (14)	0
Median (min-max)	3 (1-9)	4 (1-9)	4 (2-8)
Germline *TP53* Y220C, n (%)			
Negative	66 (99)	22 (100)	9 (100)
Positive	1 (1)	0	0
***KRAS* mutation status**			
Wildtype	50 (75)	22 (100)	9 (100)
KRAS SNV*	17 (25)	0	0
Measurable disease at baseline			
Yes	64 (95)	20 (91)	8 (89)

Data cut-off: 05 Sept 2023. *12 pancreas, three colon, one small intestine, one cholangiocarcinoma.
BID, twice daily; ECOG PS, Eastern Cooperative Oncology Group Performance Score; QD, once daily; SNV, single-nucleotide variant.

- Across the total population (n=67) treatment-related adverse events were mostly Grade 1/2 (**Table 2**)
 - Administration of rezatapopt with food led to an improvement in gastrointestinal toxicities[11]
 - The safety profile in the subgroup of patients with ovarian and breast cancers was consistent with the overall population

Table 2. Incidence of TRAEs in ≥5% of patients receiving rezatapopt (1150 mg QD to 1500 BID)

All TRAEs, n (%)		Max CTCAE			
Preferred term	**Overall n=67**	**1**	**2**	**3**	**4**
Any TRAE	**60 (89.6)**	**16 (23.9)**	**27 (40.3)**	**16* (23.9)**	**1† (1.5)**
Nausea	34 (50.7)	22 (32.8)	11 (16.4)	1 (1.5)	–
Vomiting	29 (43.3)	16 (23.9)	12 (17.9)	1 (1.5)	–
Blood creatinine increased	18 (26.9)	10 (14.9)	8 (11.9)	–	–
Diarrhea	13 (19.4)	12 (17.9)	–	1 (1.5)	–
Fatigue	13 (19.4)	8 (11.9)	5 (7.5)	–	–
ALT increased	12 (17.9)	4 (6.0)	5 (7.5)	3 (4.5)	–
AST increased	11 (16.4)	7 (10.4)	2 (3.0)	2 (3.0)	–
Anemia	10 (14.9)	1 (1.5)	6 (9.0)	3 (4.5)	–
Decreased appetite	7 (10.4)	2 (3.0)	4 (6.0)	1 (1.5)	–
Proteinuria	6 (9.0)	1 (1.5)	5 (7.5)	–	–
Rash maculo-papular	6 (9.0)	1 (1.5)	3 (4.5)	2 (3.0)	–
Headache	5 (7.5)	4 (6.0)	1 (1.5)	–	–
Lipase increased	5 (7.5)	4 (6.0)	–	1 (1.5)	–
Platelet count decreased	4 (6.0)	1 (1.5)	1 (1.5)	2 (3.0)	–
Amylase increased	4 (6.0)	3 (4.5)	1 (1.5)	–	–
Dehydration	4 (6.0)	–	4 (6.0)	–	–

Data cut-off: 05 Sept 2023. *Includes five additional Grade 3 TRAEs: neutrophil count decreased, acute kidney injury, pancreatitis, pneumonitis, and rash erythematous. Note that a patient could have multiple Grade 3 events. †Includes one patient with Grade 4 immune thrombocytopenia.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; BID, twice daily; CTCAE, Common Terminology Criteria for Adverse Events; QD, once daily; TRAE, treatment-related adverse event.

- Confirmed partial responses were observed in 13/38 (34%) efficacy-evaluable patients with solid tumors including 7/15 (47%) patients with ovarian cancer and 3/8 (38%) patients with breast cancer (**Table 3**, **Figure 1A–C**)

Table 3. Best overall response in the *TP53* Y220C/*KRAS* wild-type efficacy-evaluable population receiving rezatapopt in the efficacious dose range and the recommended phase 2 dose (2000 mg QD)

	2000 mg QD N=16	**1150 mg QD–1500 mg BID N=38**
	ORR, n (%)	**ORR, n (%)**
Overall	**6/16 (38)**	**13/38 (34)**
Confirmed PR	6/16 (38)	13/38 (34)
SD	8/16 (50)	20/38 (53)
PD	2/16 (13)	5/38 (13)
	2000 mg QD	**1150 mg QD–1500 mg BID**
	ORR, n (%)	**ORR, n (%)**
Ovary	2/5 (40)	7/15 (47)
Breast	2/3 (67)	3/8 (38)
Small cell lung	0/1 (0)	1/2 (50)
Endometrial	1/1 (100)	1/1 (100)
Other	1/6 (17)	1/12 (8)

Data cut-off: 05 Sept 2023.
BID, twice daily; ORR, overall response rate; PD, progressive disease; PR, partial response; QD, once daily; SD, stable disease.

Figure 1. Target lesion reduction in the *TP53* Y220C/*KRAS* wild-type efficacy-evaluable population receiving rezatapopt in the efficacious dose range with measurable disease for (A) the overall population across tumor types, (B) the ovarian cancer subgroup, and (C) the breast cancer subgroup*

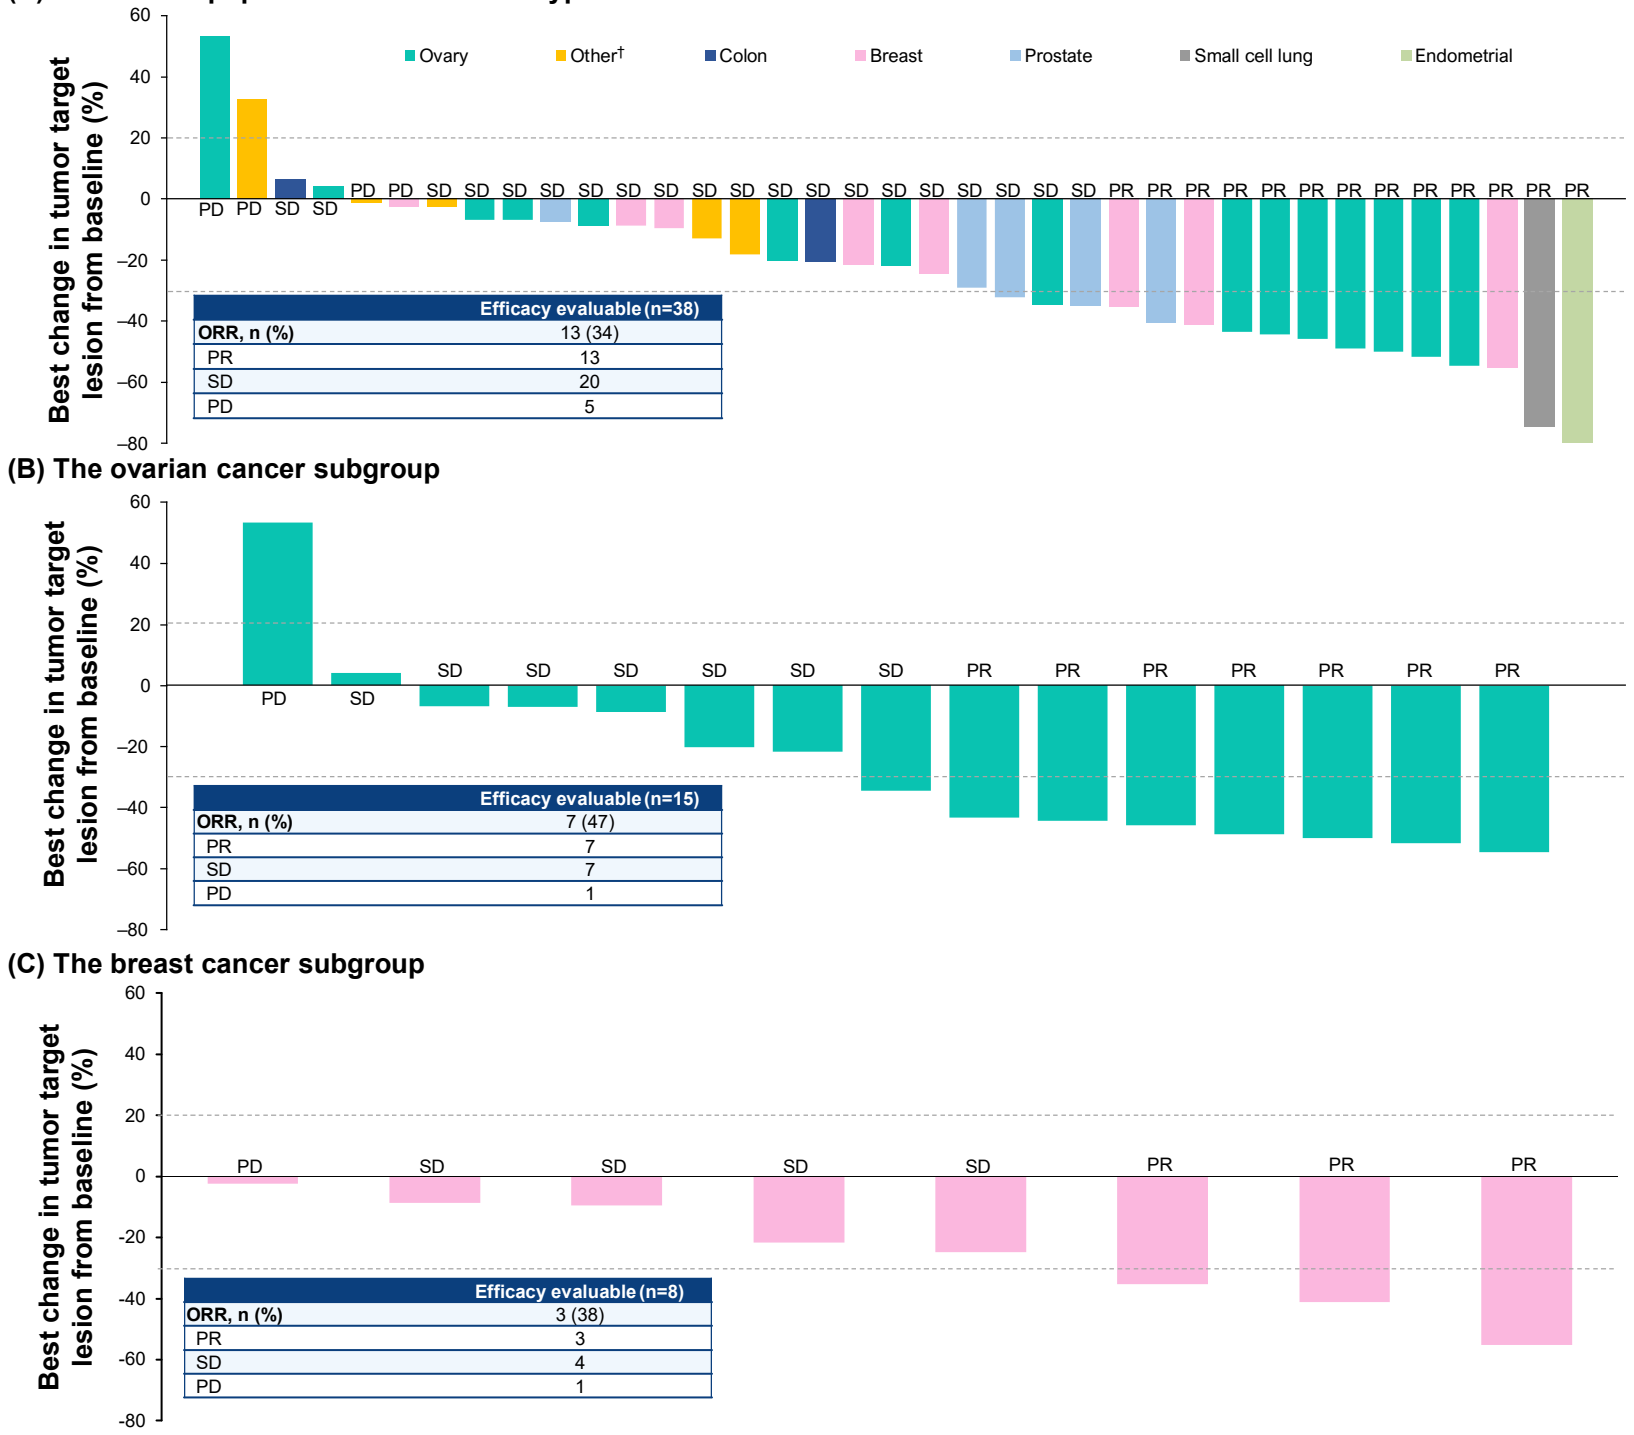
(A) The overall population across tumor types
Ovary
Other†
Colon
Breast
Prostate
Small cell lung
Endometrial
Best change in tumor target lesion from baseline (%)
Efficacy evaluable (n=38)
ORR, n (%) 13 (34)
PR 13
SD 20
PD 5
(B) The ovarian cancer subgroup
Best change in tumor target lesion from baseline (%)
PD SD SD SD SD SD SD SD PR PR PR PR PR PR PR
Efficacy evaluable (n=15)
ORR, n (%) 7 (47)
PR 7
SD 7
PD 1
(C) The breast cancer subgroup
Best change in tumor target lesion from baseline (%)
PD SD SD SD SD PR PR PR
Efficacy evaluable (n=8)
ORR, n (%) 3 (38)
PR 3
SD 4
PD 1

Data cut-off: 05 Sept 2023. *Includes patients with measurable disease at baseline and ≥1 post-baseline tumor assessment. One patient with small cell lung cancer without tumor measurement at first scan is not represented. †Other tumor types include sarcoma, esophageal cancer, germ cell tumor, pleomorphic rhabdomyosarcoma, and urothelial cancer.
ORR, overall response rate; PD, progressive disease; PR, partial response; SD, stable disease.

- The PYNNACLE registrational Phase 2 study is an ongoing, global, pivotal, basket, single-arm, open-label, multicenter, dose-expansion trial assessing the efficacy, safety, pharmacokinetics and patient-reported outcomes of rezatapopt in patients with solid tumors (including ovarian, breast, and other cancers) that are *TP53* Y220C mutated and *KRAS* wild type (**Figure 2**)[9,10,11]
- Eligible patients with solid tumors are receiving rezatapopt at the recommended Phase 2 dose of 2000 mg orally QD with food for continuous 21-day cycles; patients will be followed until lost to follow-up, death, 2 years after last patient discontinuation, or end of study[9,10,12]

Figure 2. PYNNACLE Phase 2 study design[9,10,11]

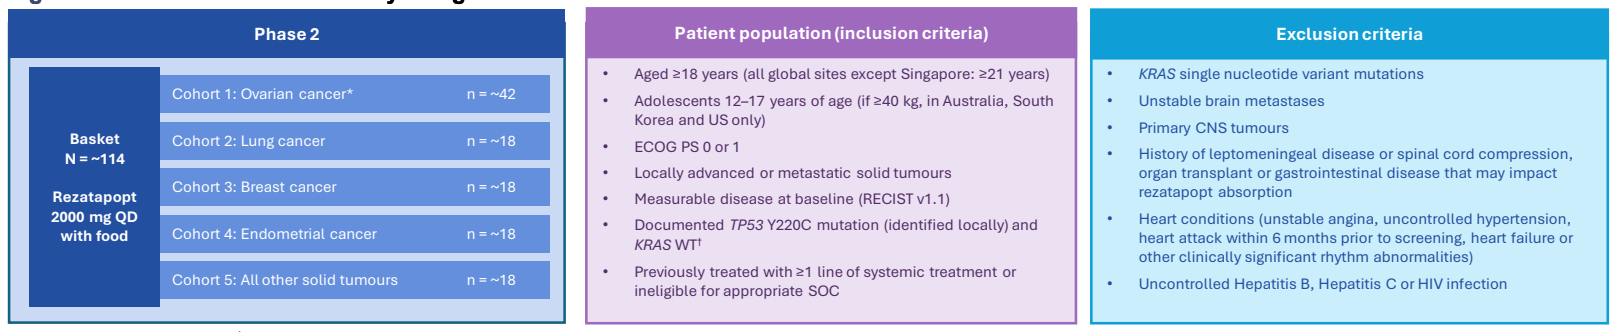
Phase 2
Basket N = ~114
Rezatapopt 2000 mg QD with food
Cohort 1: Ovarian cancer* n = ~42
Cohort 2: Lung cancer n = ~18
Cohort 3: Breast cancer n = ~18
Cohort 4: Endometrial cancer n = ~18
Cohort 5: All other solid tumours n = ~18
Patient population (inclusion criteria)
Aged ≥18 years (all global sites except Singapore: ≥21 years)
Adolescents 12–17 years of age (if ≥40 kg, in Australia, South Korea and US only)
ECOG PS 0 or 1
Locally advanced or metastatic solid tumours
Measurable disease at baseline (RECIST v1.1)
Documented TP53 Y220C mutation (identified locally) and KRAS WT†
Previously treated with ≥1 line of systemic treatment or ineligible for appropriate SOC
Exclusion criteria
KRAS single nucleotide variant mutations
Unstable brain metastases
Primary CNS tumours
History of leptomeningeal disease or spinal cord compression, organ transplant or gastrointestinal disease that may impact rezatapopt absorption
Heart conditions (unstable angina, uncontrolled hypertension, heart attack within 6 months prior to screening, heart failure or other clinically significant rhythm abnormalities)
Uncontrolled Hepatitis B, Hepatitis C or HIV infection

*Platinum resistant or refractory. †*KRAS* WT defined as the absence of *KRAS* SNV mutations.
CNS, central nervous system; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HIV, human immunodeficiency virus; QD, once daily; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SOC, standard of care; SNV, single-nucleotide variant; WT, wild type.

CONCLUSIONS

- In the PYNNACLE Phase 1 study, rezatapopt demonstrated a favorable safety profile and promising single-agent efficacy across the efficacious dose range in heavily pre-treated patients with various solid tumors, including patients with locally advanced or metastatic ovarian and breast cancers
- The PYNNACLE Phase 2 trial is ongoing and is assessing rezatapopt as a monotherapy at the recommended Phase 2 dose of 2000 mg QD taken with food in patients with locally advanced or metastatic *TP53* Y220C-mutated and *KRAS* wild-type solid tumors
 - Locations for the PYNNACLE Phase 2 study in Australia include Perth, Adelaide, Melbourne, and Sydney[9,10]

References
1. Levine AJ. *Nat Rev Cancer*. 2020;20:471–480; 2. FoundationInsights™. A proprietary database used under license with review and approval from Foundation Medicine®. Available at: https://www.foundationmedicine.com/service/genomic-data-solutions (accessed Jul 2024); 3. Schram AM, et al. SGO Annual Meeting on Women's Cancer. 2024; Oral presentation, late-breaking abstract; 4. Dumbrava EE, et al. SABCS. 2024; Poster 3-12-27, abstract SESS-2137; 5. Soussi T, et al. *Review Cell Death Differ*. 2015;22:1239–49; 6. Binh V, et al. *ACS Med Chem Lett*. 2024; epub ahead of print; 7. Schram AM, et al. AACR-NCI-EORTC Molecular Targets and Cancer Therapeutics. 2023; Oral presentation, late-breaking abstract LB_A25; 8. Dumbrava EE, et al. ASCO. 2022; Oral presentation; 9. NCT04585750. ClinicalTrials.gov. Available at: https://clinicaltrials.gov/study/NCT04585750 (accessed Dec 2024); 10. PYNNACLE Clinical Study. Available at: https://www.pynnaclestudy.com/PC14586/ (accessed Dec 2024); 11. Kuo HCD, et al. ACCP. 2024; Poster 044, abstract 1773065; 12. Schram AM, et al. ESMO. 2024; Poster 691 TiP.

Acknowledgments
We would like to thank: All the patients, their families, and caregivers who have participated, and continue to participate, in the clinical trials; investigators and research staff; Pharmaceutical Product Development; and Resolution Biosciences and Foundation Medicine. This study and the clinical trials are sponsored by PMV Pharmaceuticals, Inc. Medical writing was provided by Carolyn Maskin and Danielle Lindley of Nucleus Global, an Inizio company, funded by PMV Pharmaceuticals, Inc.

Disclosures
MLJ: received research funding from PMV Pharmaceuticals, Inc. AWT: locally PI, institutional, financial interest, trial funding, and patents. JAT: received research funding from PMV Pharmaceuticals, Inc. SK: local PI, institutional, financial interest, trial funding. KLD, MF: PMV Pharmaceuticals, Inc. employees (with stock options). AMS: attended advisory boards and received research funding from PMV Pharmaceuticals, Inc. EED: received research funding/grant from, attended advisory boards for, and provided speaker role for PMV Pharmaceuticals, Inc. All other authors declare no conflicts.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from APOS and the author of this poster

Scan QR code to download the poster and for author disclosures

For further information please contact: clinicaltrials@pmvpharma.com

MA-586-0054 March 2025

Presented at the Australian Precision Oncology Symposium 2025, March 7–8, 2025, Kensington, NSW, Australia

Presented by: Amy Body

Locations for the PYNNACLE Phase 2 study in Australia

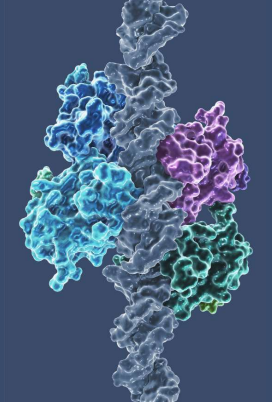

Perth
Location: Linear Clinical Research
Nedlands WA 6009, Australia
Contact: Michael Millward, MD
Status: Recruiting

Melbourne
Location: Monash Medical Centre
Clayton VIC 3168, Australia
Contact: Amy Body, MD
Status: Recruiting

Adelaide
Location: Flinders Medical Centre
Bedford Park SA 5042, Australia
Contact: Shwagi Sukumaran, MD
Status: Recruiting soon

Sydney
Location: Chris O’Brien
Lifehouse Hospital
Camperdown NSW 2050, Australia
Contact: Peter Grimison, MD
Status: Recruiting