Small Molecule Reactivators of Y220C Mutant p53 Modulate Tumor Infiltrating Leukocytes and Synergize with Immune Checkpoint Inhibitors

Anna Puzio-Kuter, Chris Mulligan, Brandon Russo, Amy Wiebesiek, Lizhong Xu, Hong Yang, Binh Vu, and Melissa Dumble

2659

Abstract

Half of human cancers possess mutations in the TP53 gene, with most clustering as hotspots in the DNA binding domain. A tyrosine to cysteine substitution at amino acid 220 of the p53 protein (Y220C) is one such hotspot mutation (~1 % of all solid tumors). Small molecules that reactivate Y220C mutant p53 activity to be like wild type have been developed at PMV Pharmaceuticals, and the lead compound PC14586 is in Phase I clinical development. The molecules were designed to bind tightly to a crevice within the Y220C p53 mutant protein and stabilize it in the wild type conformation allowing reactivation of p53 transcriptional activity and expression of target proteins (e.g. p21, MDM2), resulting in tumor regression in Y220C p53 mutant xenograft models grown in immunocompromised mice.

p53 has been documented to contribute to immune responses by activating regulators of immune signaling pathways and to result in T-cell exclusion in tumor models. To investigate the role of p53 and the immune system in tumorigenesis, a Y220C human p53 knock-in (HUPKI) mouse was generated. Mice that are homozygous mutant for the Y220C humanized alleles succumb to lymphomas and sarcomas within 6 months, and cell lines generated from these tumors were shown to be sensitive in an in vitro proliferation assay to Y220C p53 reactivators (IC_{50} ~ 192-722 nM) and used to generate Y220C mutant syngeneic mouse models.

Administration of Y220C p53 reactivators in Y220C p53 syngeneic mouse tumor models resulted in a dose responsive anti-tumor effect, with maximally efficacious doses resulting in durable cures in nearly all animals, more than observed in immunocompromised mouse models. Addition of checkpoint agents with sub-efficacious administration of Y220C p53 reactivating compounds resulted in synergistic increases in mean survival time of mice, longer than with either single agent. Immunophenotyping analysis of HUPKI Y220C p53 mutant tumors exposed to p53 reactivator compounds showed modulation of the tumor immune environment. Changes include dose responsive increases in T-cells ($CD4^+$, $CD8^+$), T-regulatory cells, natural killer T (NKT) cells and dose responsive decreases in macrophages (M2) and g-MDSC cells. The synergy observed with an anti-PD-1 combination appears driven by an increase in $CD8^+$ T cells, given a decrease in efficacy in mice depleted of $CD8^+$ T cells, following exposure to anti-CD8. Further investigation into the role of Y220C p53 reactivation in immune modulation showed an increase in a Tumor Inflammation Signature (nanoString) in a dose and time dependent manner. Taken together, these data suggest a role for p53 signaling in encouraging an immunologically hot tumor microenvironment and provide support for testing small molecule reactivators of mutant p53 in combination with immune checkpoint inhibitors.

Biological Background of Y220C p53 Reactivators

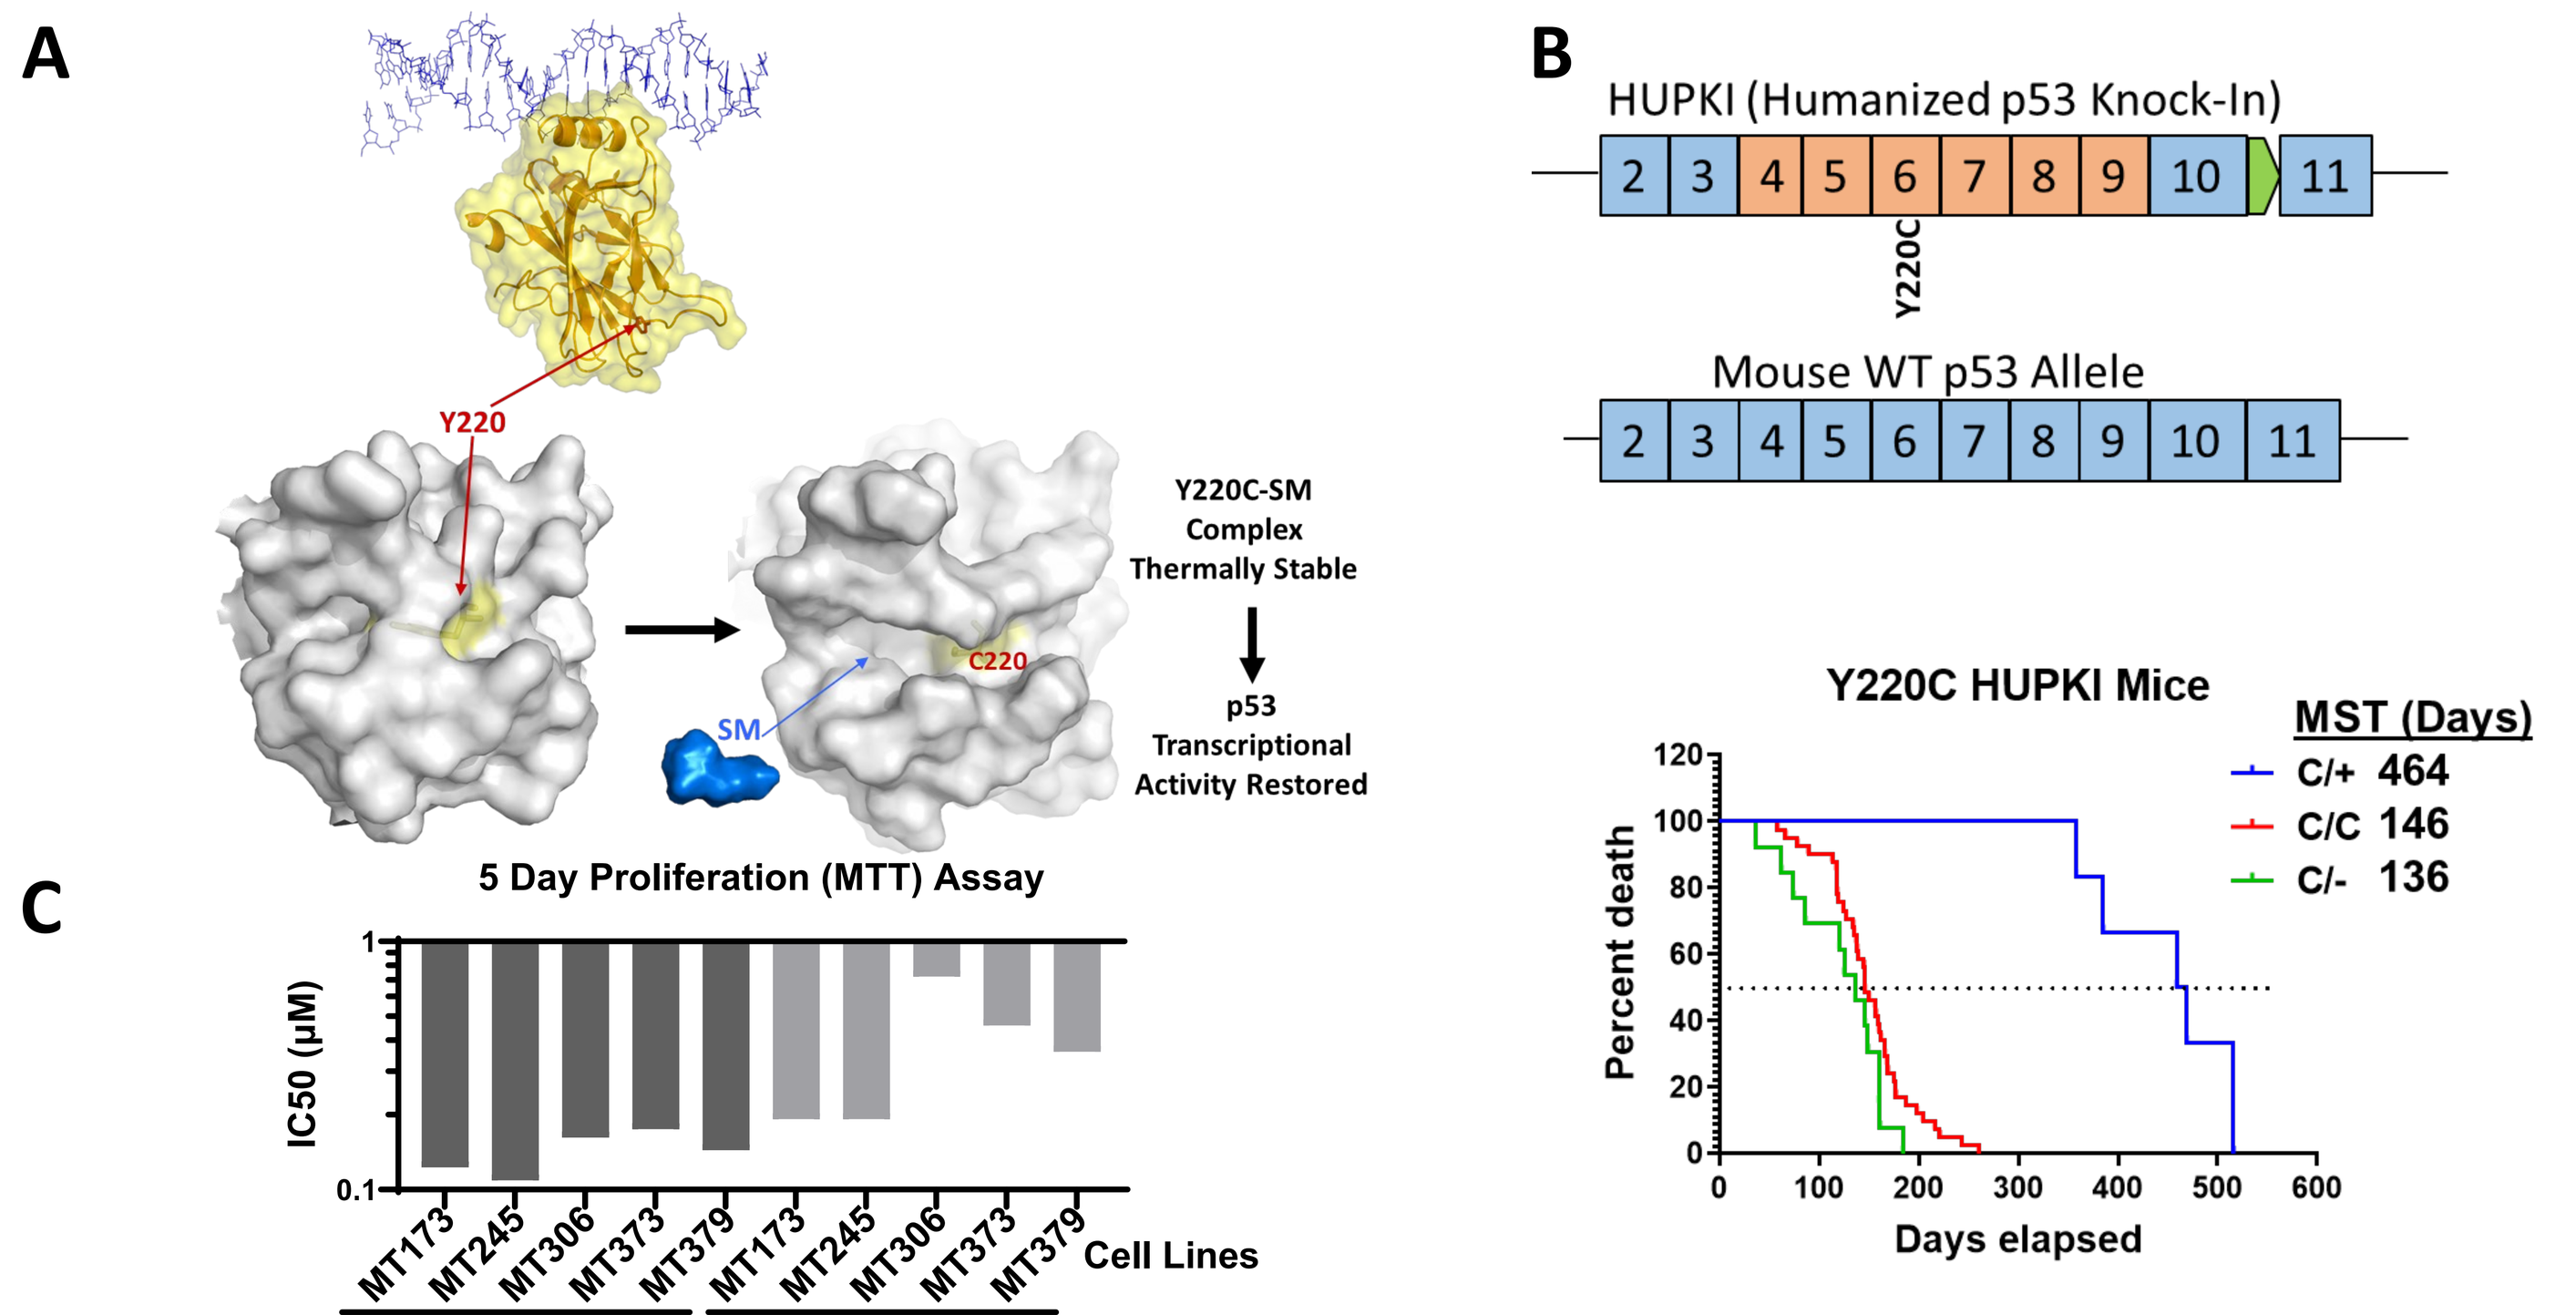
A
Y220
C220
SM
Y220C-SM Complex Thermally Stable
p53 Transcriptional Activity Restored
B
HUPKI (Humanized p53 Knock-In)
Y220C
Mouse WT p53 Allele
Y220C HUPKI Mice
MST (Days)
C/+ 464
C/C 146
C/- 136
Percent death
Days elapsed
C
5 Day Proliferation (MTT) Assay
IC50 (µM)
Cell Lines
PC14374
PC14586

FIGURE 1:

A. p53 reactivators are small molecules designed to bind to and stabilize Y220C mutant p53 in a wildtype conformation. The single amino acid change (Tyr to Cys) creates a crevice in the mutant p53 protein, making it thermally unstable and unable to effectively interact with DNA. By selectively binding this crevice, the small molecule restores WT p53 structure and reactivates its function. Two closely related Y220C p53 reactivators, PC14374 and PC14586, were used for these studies.

B. Human Y220C p53 knock-in (i.e. HUPKI) mice were generated in a C57Bl/6 background where the human binding domain (exons 4-9) was knocked-in to the mouse TP53 gene containing the Y220C mutation. Mice homozygous for the Y220C p53 mutation (C/C), or heterozygous with one Y220C mutant allele and a null allele (C/-) show a decreased survival compared to mice heterozygous for the Y220C mutation (C/+). Mice succumb to lymphomas and sarcomas resulting in decreased survival. Median survival time (MST).

C. Several lymphoma and sarcoma cell lines were generated from HUPKI-Y220C mouse tumors and were sensitive to p53 reactivators with IC_{50} value ranges of 0.109-0.175 µM for PC14374 and 0.192-0.722 µM for PC14586 in a 5 day MTT assay. Two sarcoma lines (MT245 and MT373) were shown to grow optimally in vivo and were chosen for efficacy and PD studies.

Y220C p53 Reactivators Synergize with Anti-PD-1

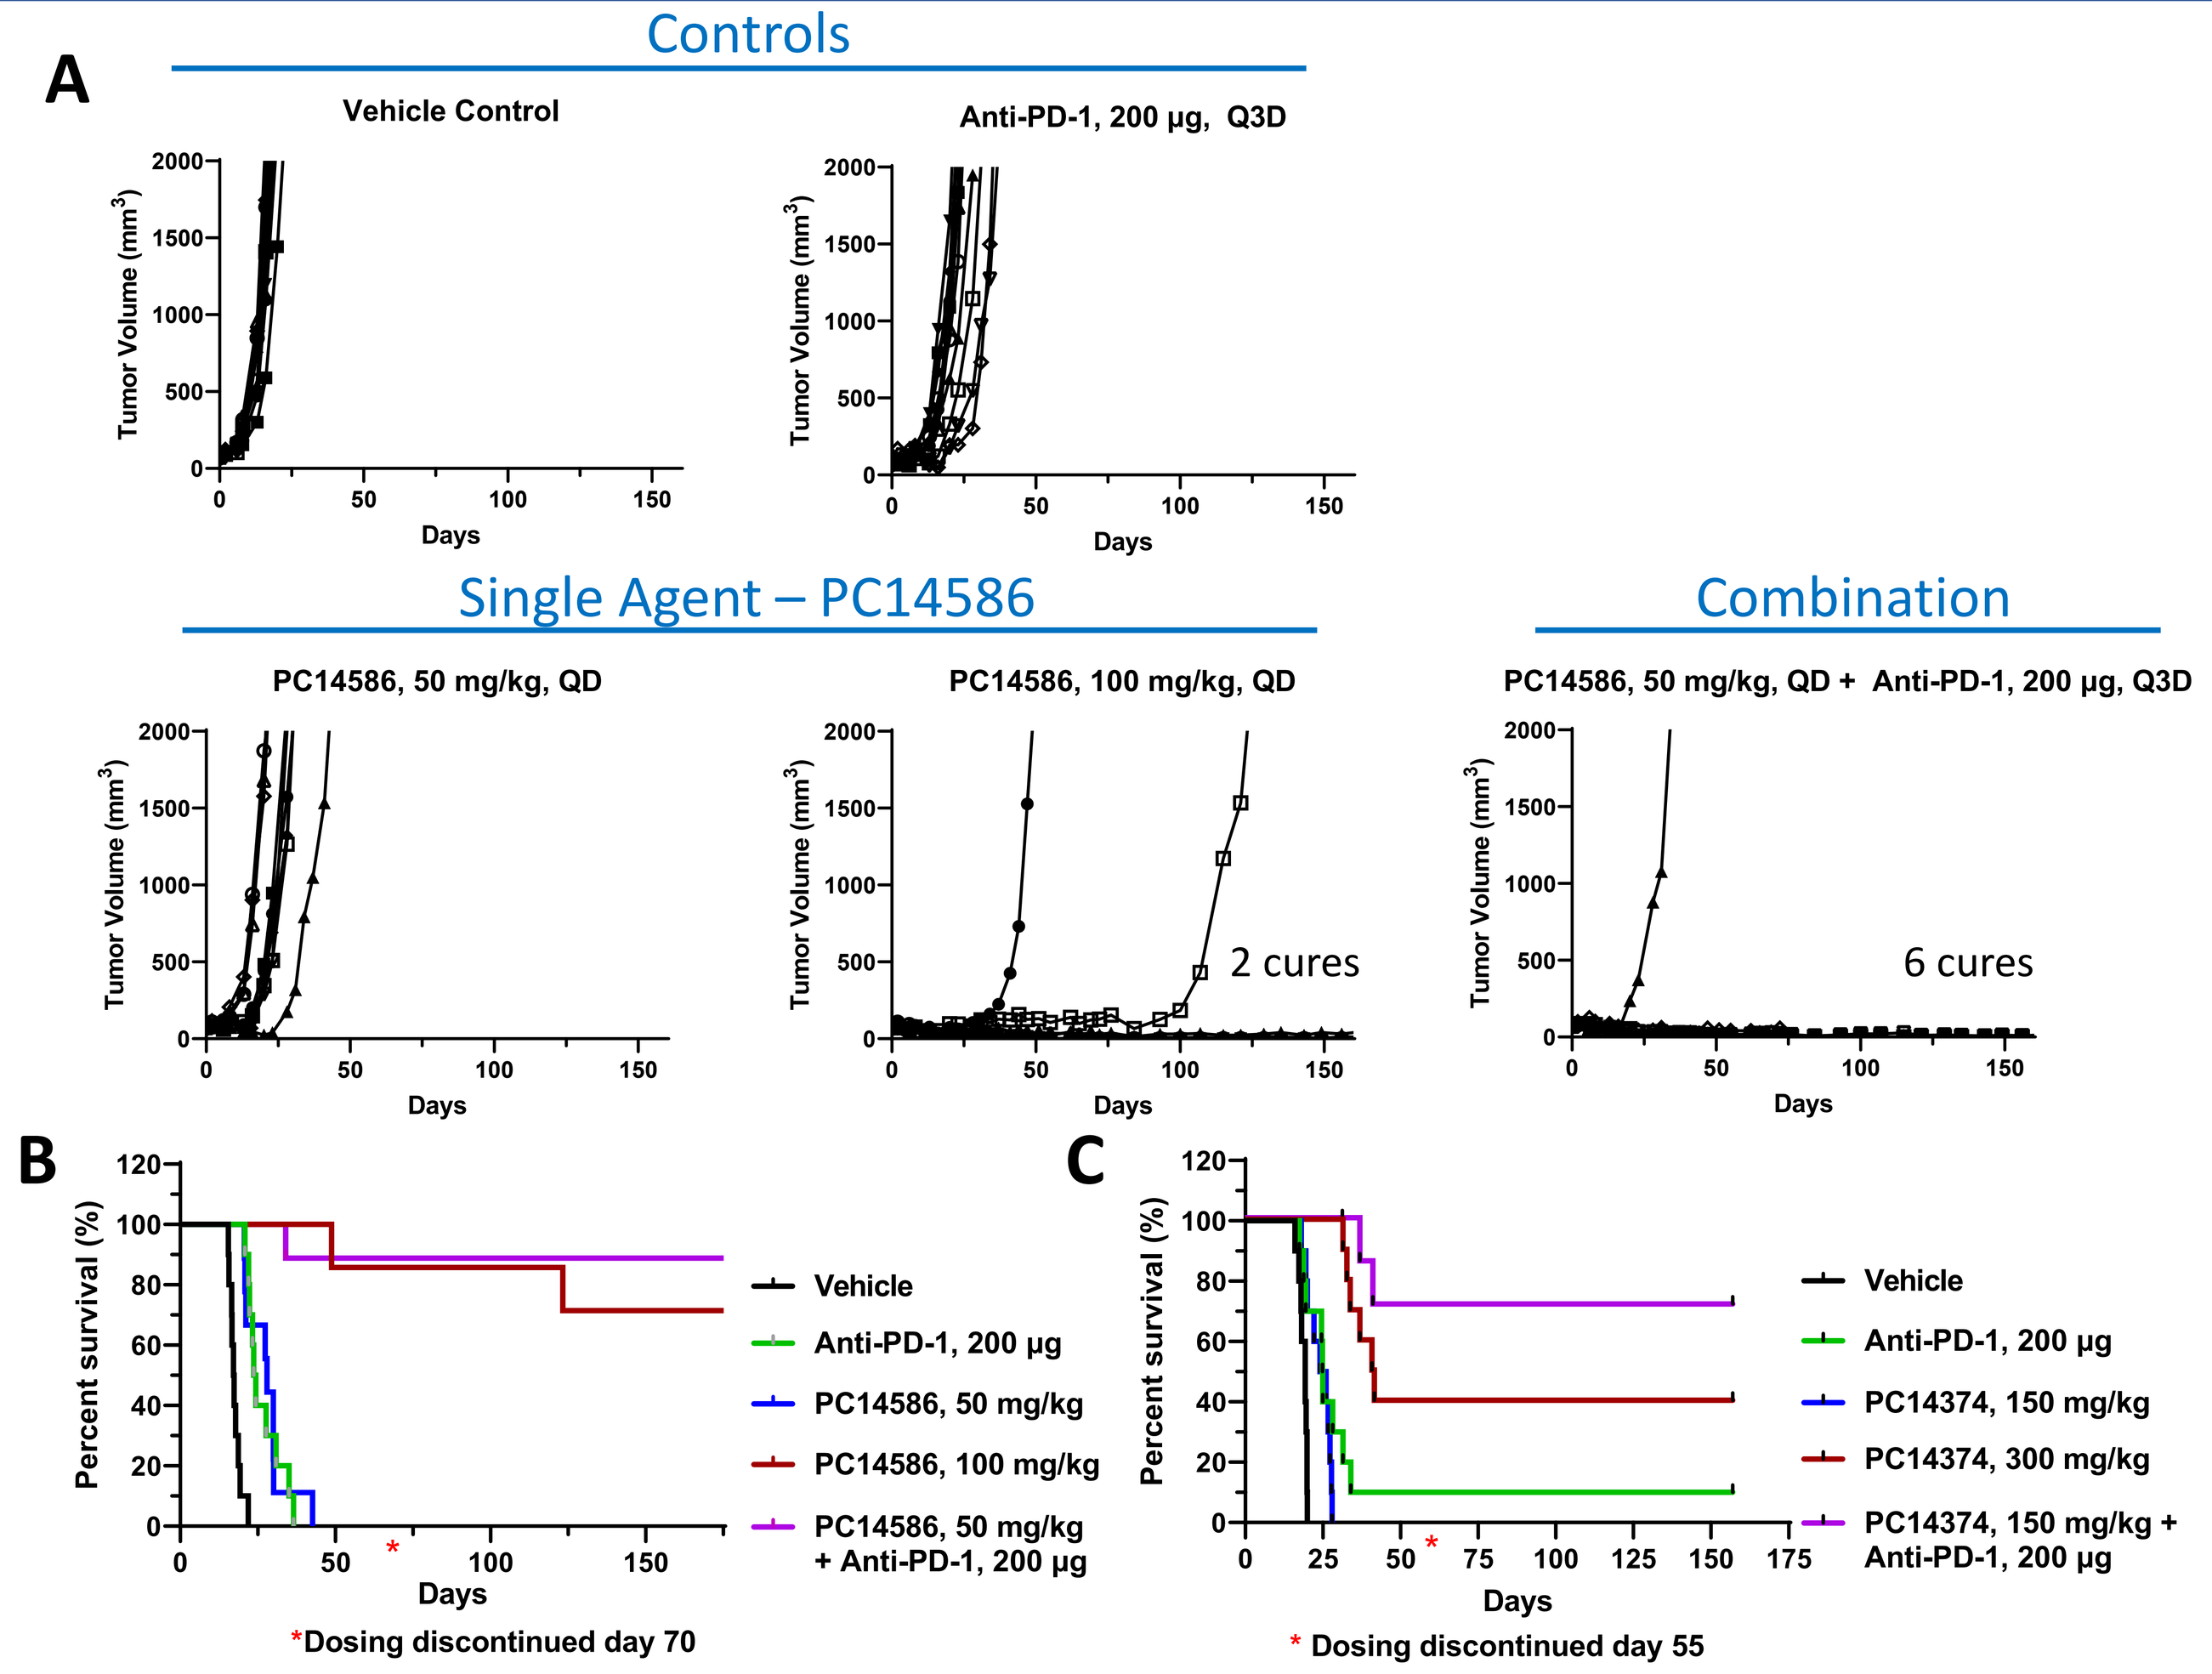
A
Controls
Vehicle Control
Anti-PD-1, 200 µg, Q3D
Single Agent – PC14586
PC14586, 50 mg/kg, QD
PC14586, 100 mg/kg, QD
2 cures
Combination
PC14586, 50 mg/kg, QD + Anti-PD-1, 200 µg, Q3D
6 cures
Tumor Volume (mm³)
Days
B
Percent survival (%)
Vehicle
Anti-PD-1, 200 µg
PC14586, 50 mg/kg
PC14586, 100 mg/kg
PC14586, 50 mg/kg + Anti-PD-1, 200 µg
*Dosing discontinued day 70
C
Vehicle
Anti-PD-1, 200 µg
PC14374, 150 mg/kg
PC14374, 300 mg/kg
PC14374, 150 mg/kg + Anti-PD-1, 200 µg
* Dosing discontinued day 55

Group	MST (Days)
Vehicle	21
Anti-PD-1, 200 µg, Q3D	24
PC14586, 50 mg/kg, QD	28
PC14586, 100 mg/kg, QD	>182
PC14586, 50 mg/kg, QD + Anti-PD-1, 200 µg, Q3D	>182

Group	MST (Days)
Vehicle	19
Anti-PD-1, 200 µg, Q3D	25
PC14374, 150 mg/kg, Q7D	25
PC14374, 300 mg/kg, Q7D	44
PC14374, 150 mg/kg, Q7D + Anti-PD-1, 200 µg Q3D	>157

FIGURE 2:
HUPKI Y220C p53 mutant sarcoma cell lines (Figure 1) were implanted subcutaneously into C57Bl/6 mice. Day 6 post implantation mice were randomized into treatment groups (n=10/group) according to tumor volume. Individual mouse tumor growth curves volumes show anti-tumor activity and anti-PD-1 synergy with administration of (A) PC14586 at 50 or 100 mg/kg QD, PO as single agents or in combination with anti-PD-1 at 200 µg, IP (Clone RMPI-14 BioXCell). A study with PC14374 at 150 or 300 mg/kg Q7D (oral, once daily, once per week) showed similar results. Kaplan-Meier curves show a significant increase in median survival time (MST) with the combination of either p53 reactivator, PC14586 (B) or PC14374 (C), with anti-PD-1. After actively growing tumors reached survival endpoint and dosing was discontinued, "durable" cures were monitored to day 182 (PC14586 study) and day 157 (PC14374 study).

Transient Conversion of Mutant Y220C to WT p53 with Small Molecule p53 Reactivators

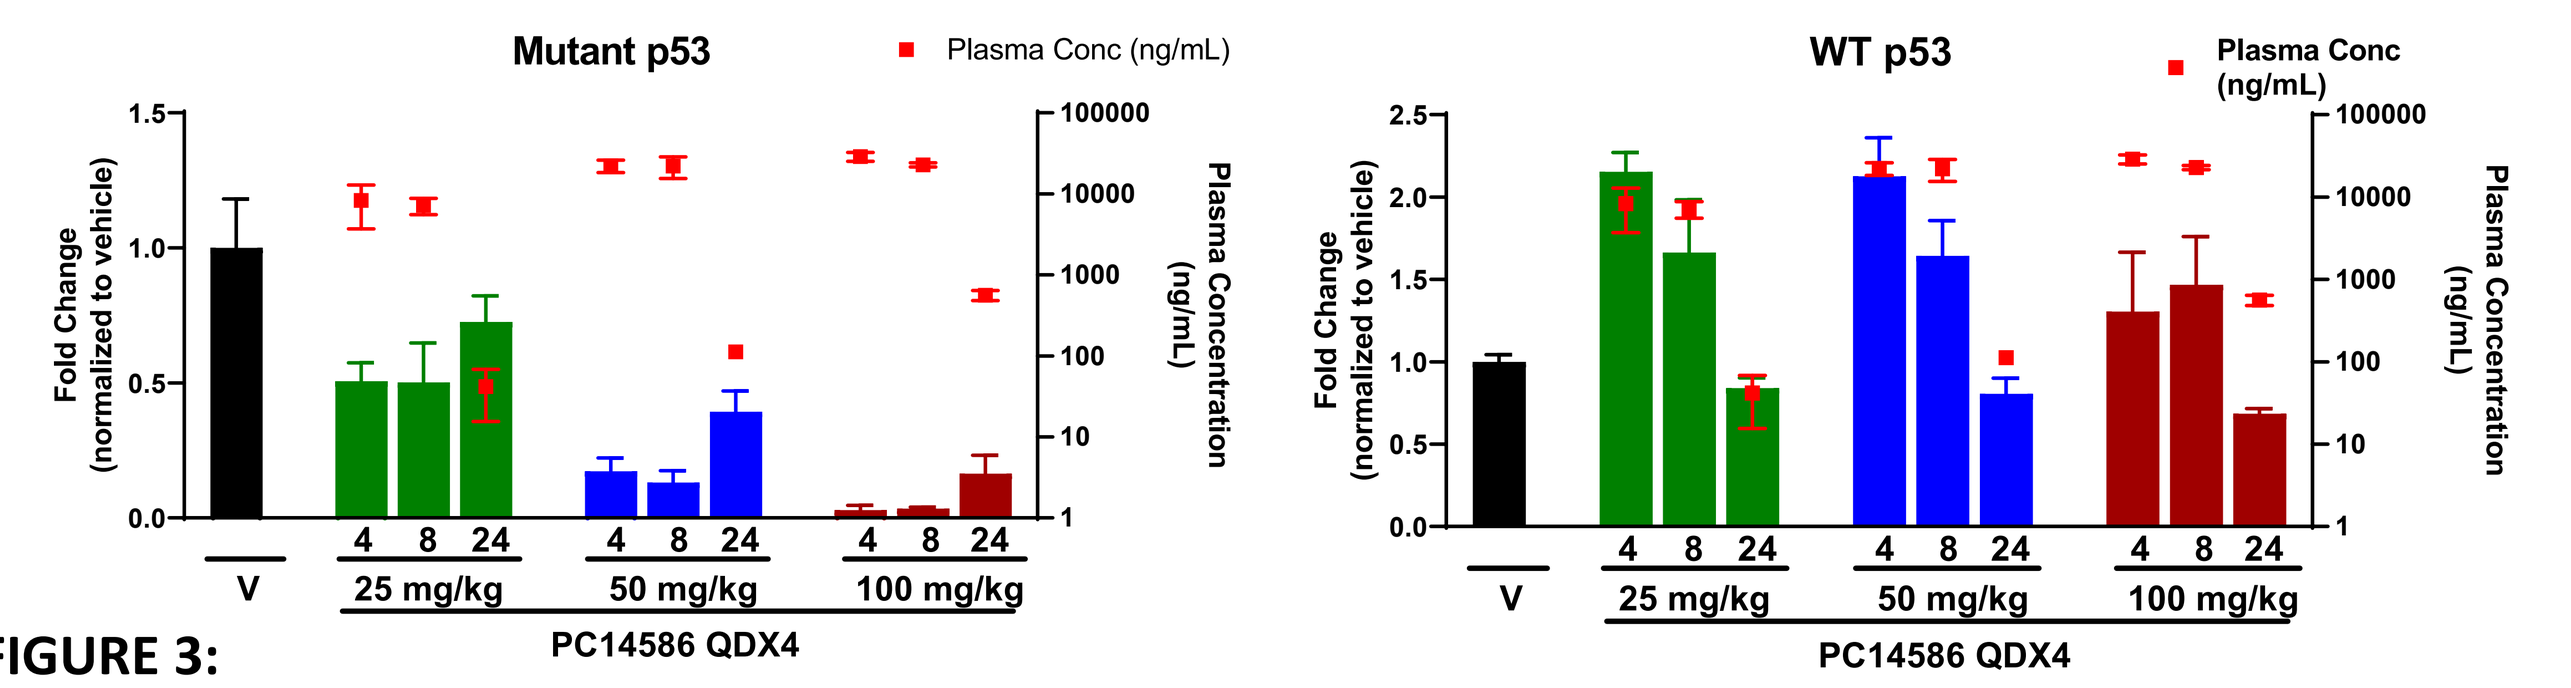
Mutant p53
Plasma Conc (ng/mL)
WT p53
Fold Change (normalized to vehicle)
Plasma Concentration (ng/mL)
25 mg/kg
50 mg/kg
100 mg/kg
PC14586 QDX4

FIGURE 3:
Tumors from mice administered PC14586 at 25, 50 and 100 mg/kg QDx4 demonstrated a reduction in mutant p53 protein levels at 4, 8 and 24 h post-dose and an increase in wild-type p53 protein levels 4 and 8 h post-dose compared to control treated tumors. The reduction of mutant p53 and increase in WT p53 levels correspond to peak plasma concentration (~10,000 ng/mL) of PC14586. A PD study with PC14374 at 75 and 150 mg/kg 2QDx1 showed similar results with decreases in mutant p53 levels and increase in WT p53 levels (data not shown).

Y220C p53 Reactivation Results in Tumor T-Cell Increases and Macrophage, g-MDSC Decreases

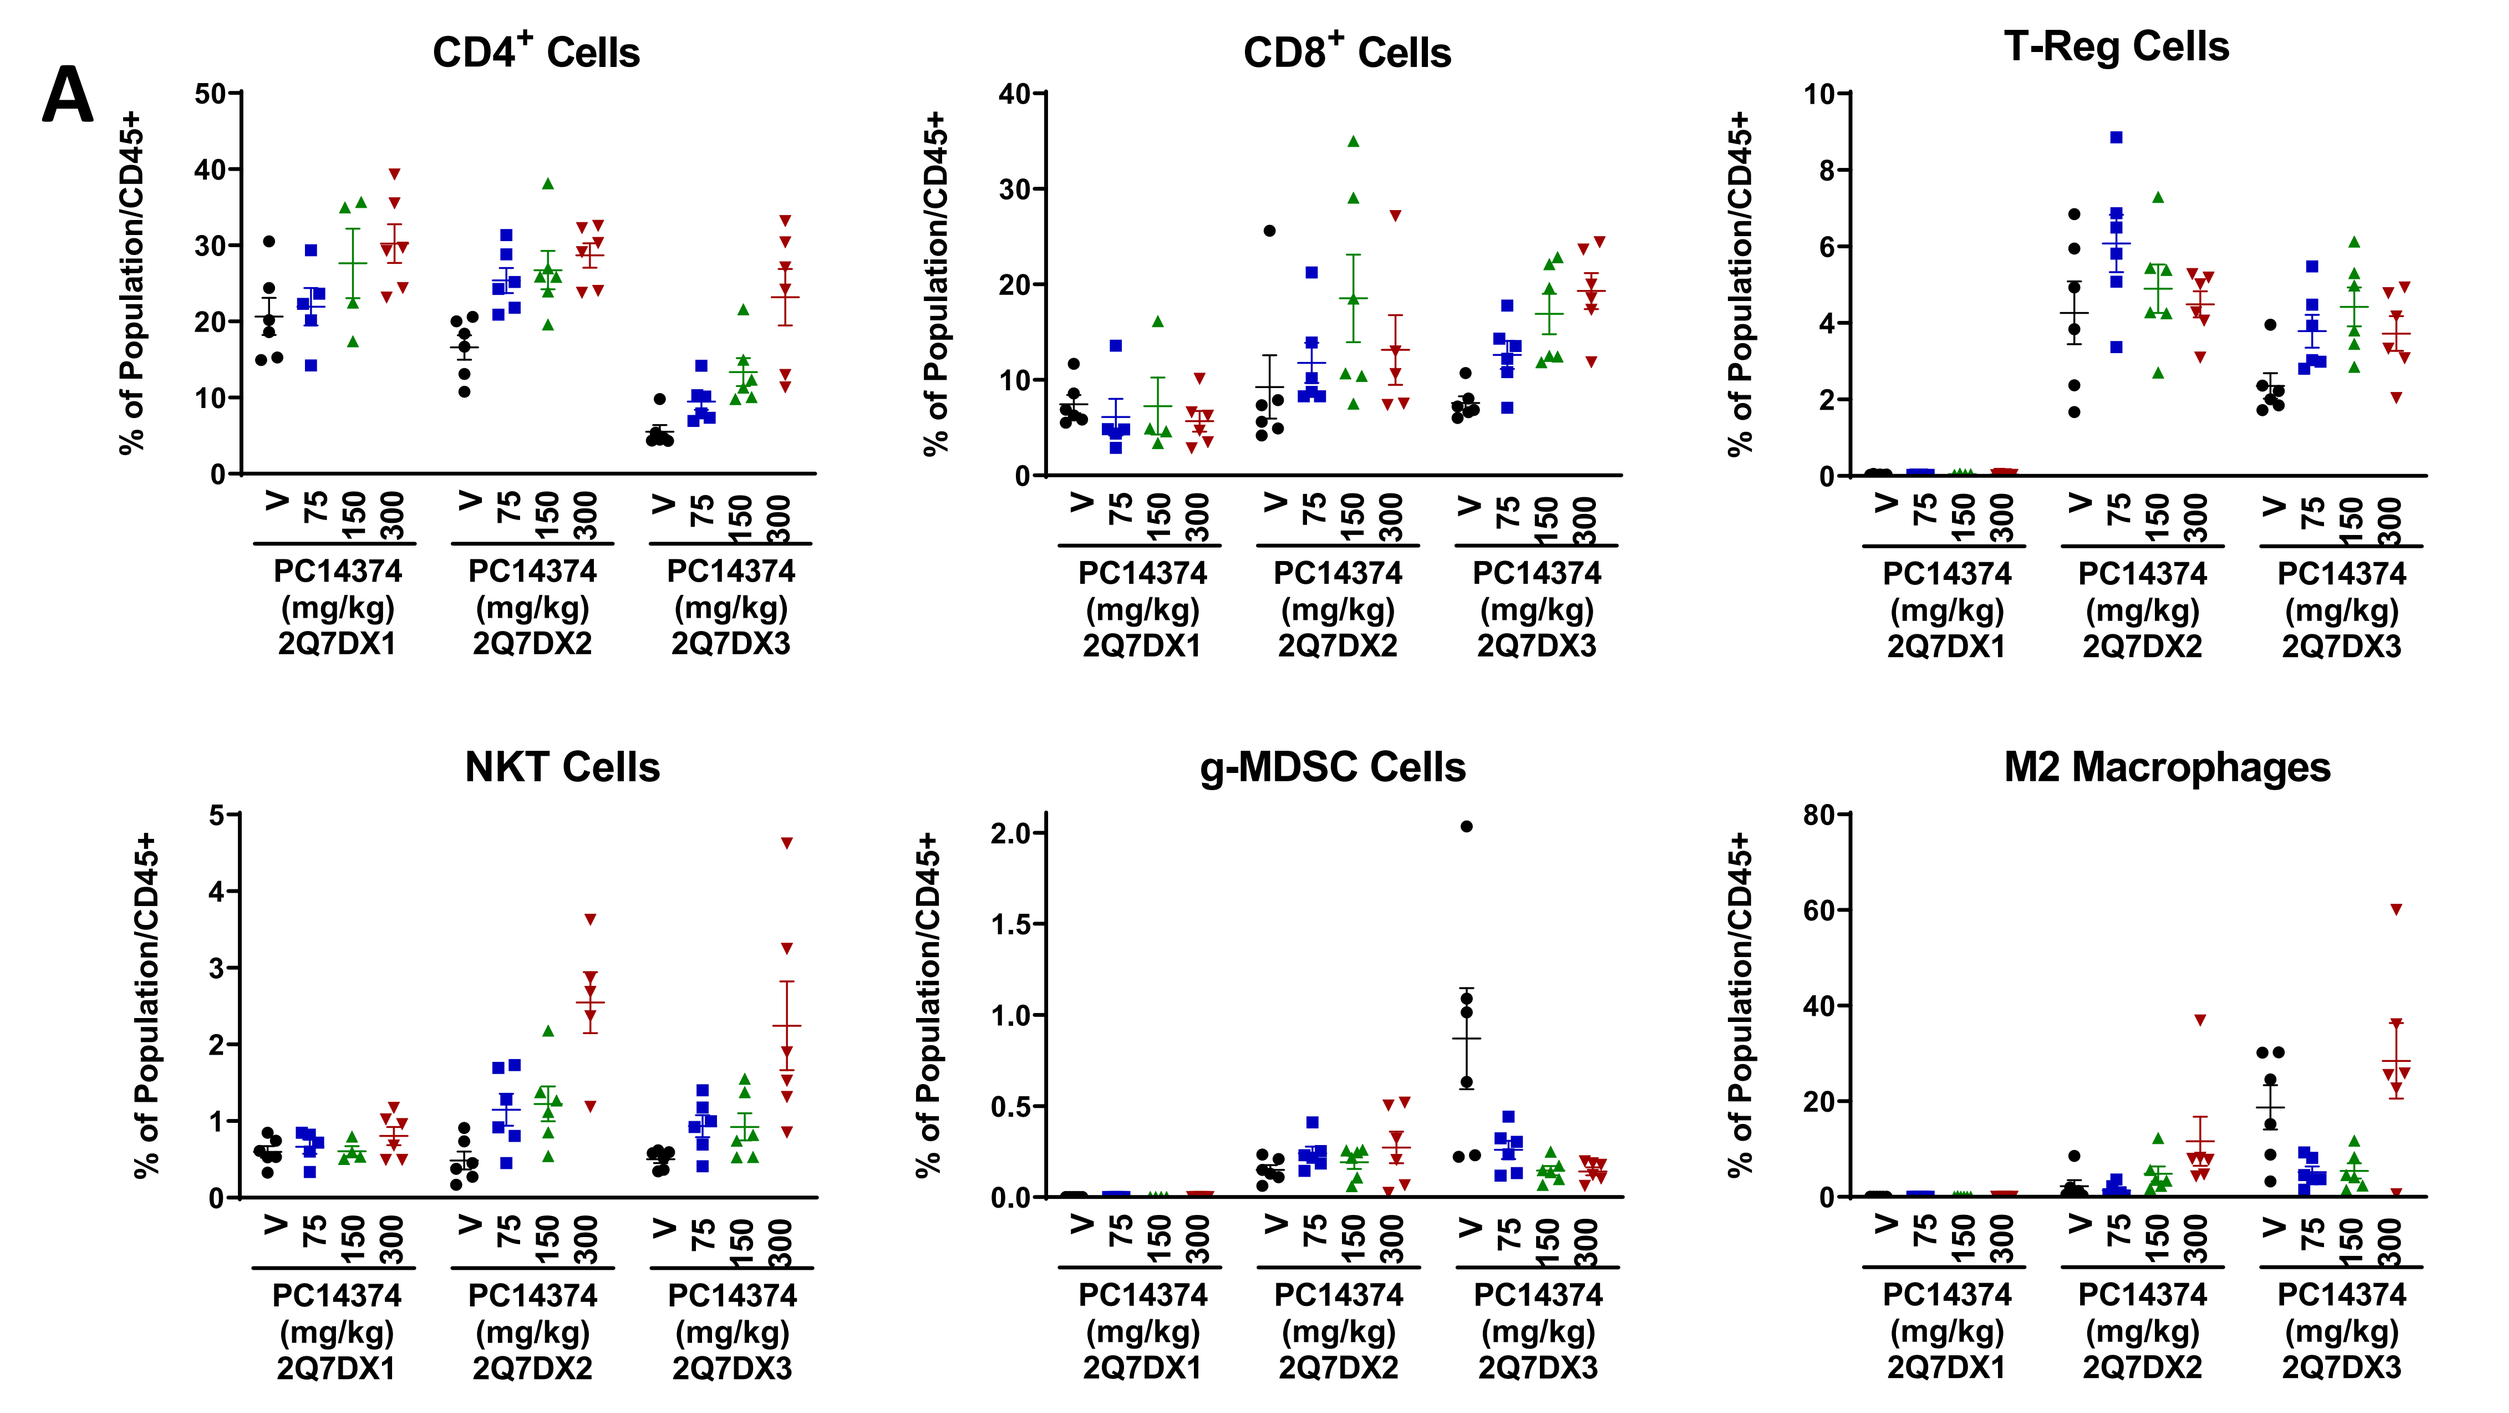
A
CD4+ Cells
CD8+ Cells
T-Reg Cells
NKT Cells
g-MDSC Cells
M2 Macrophages
% of Population/CD45+
PC14374 (mg/kg) 2Q7DX1
PC14374 (mg/kg) 2Q7DX2
PC14374 (mg/kg) 2Q7DX3

FIGURE 4:
A. Six days post inoculation with MT373 cells, mice were administered PC14374 orally at 75, 150 or 300 mg/kg for 2Q7Dx1, 2Q7Dx2, or 2Q7Dx3 and tumors harvested 72 h post dose for analysis of tumor infiltrating lymphocytes by flow cytometry. Reactivation of p53 resulted in increases in $CD4^+$, $CD8^+$, T-Regulatory T-cells and NKT cells and decreases in g-MDSC and M2 macrophages that were dose and time dependent.

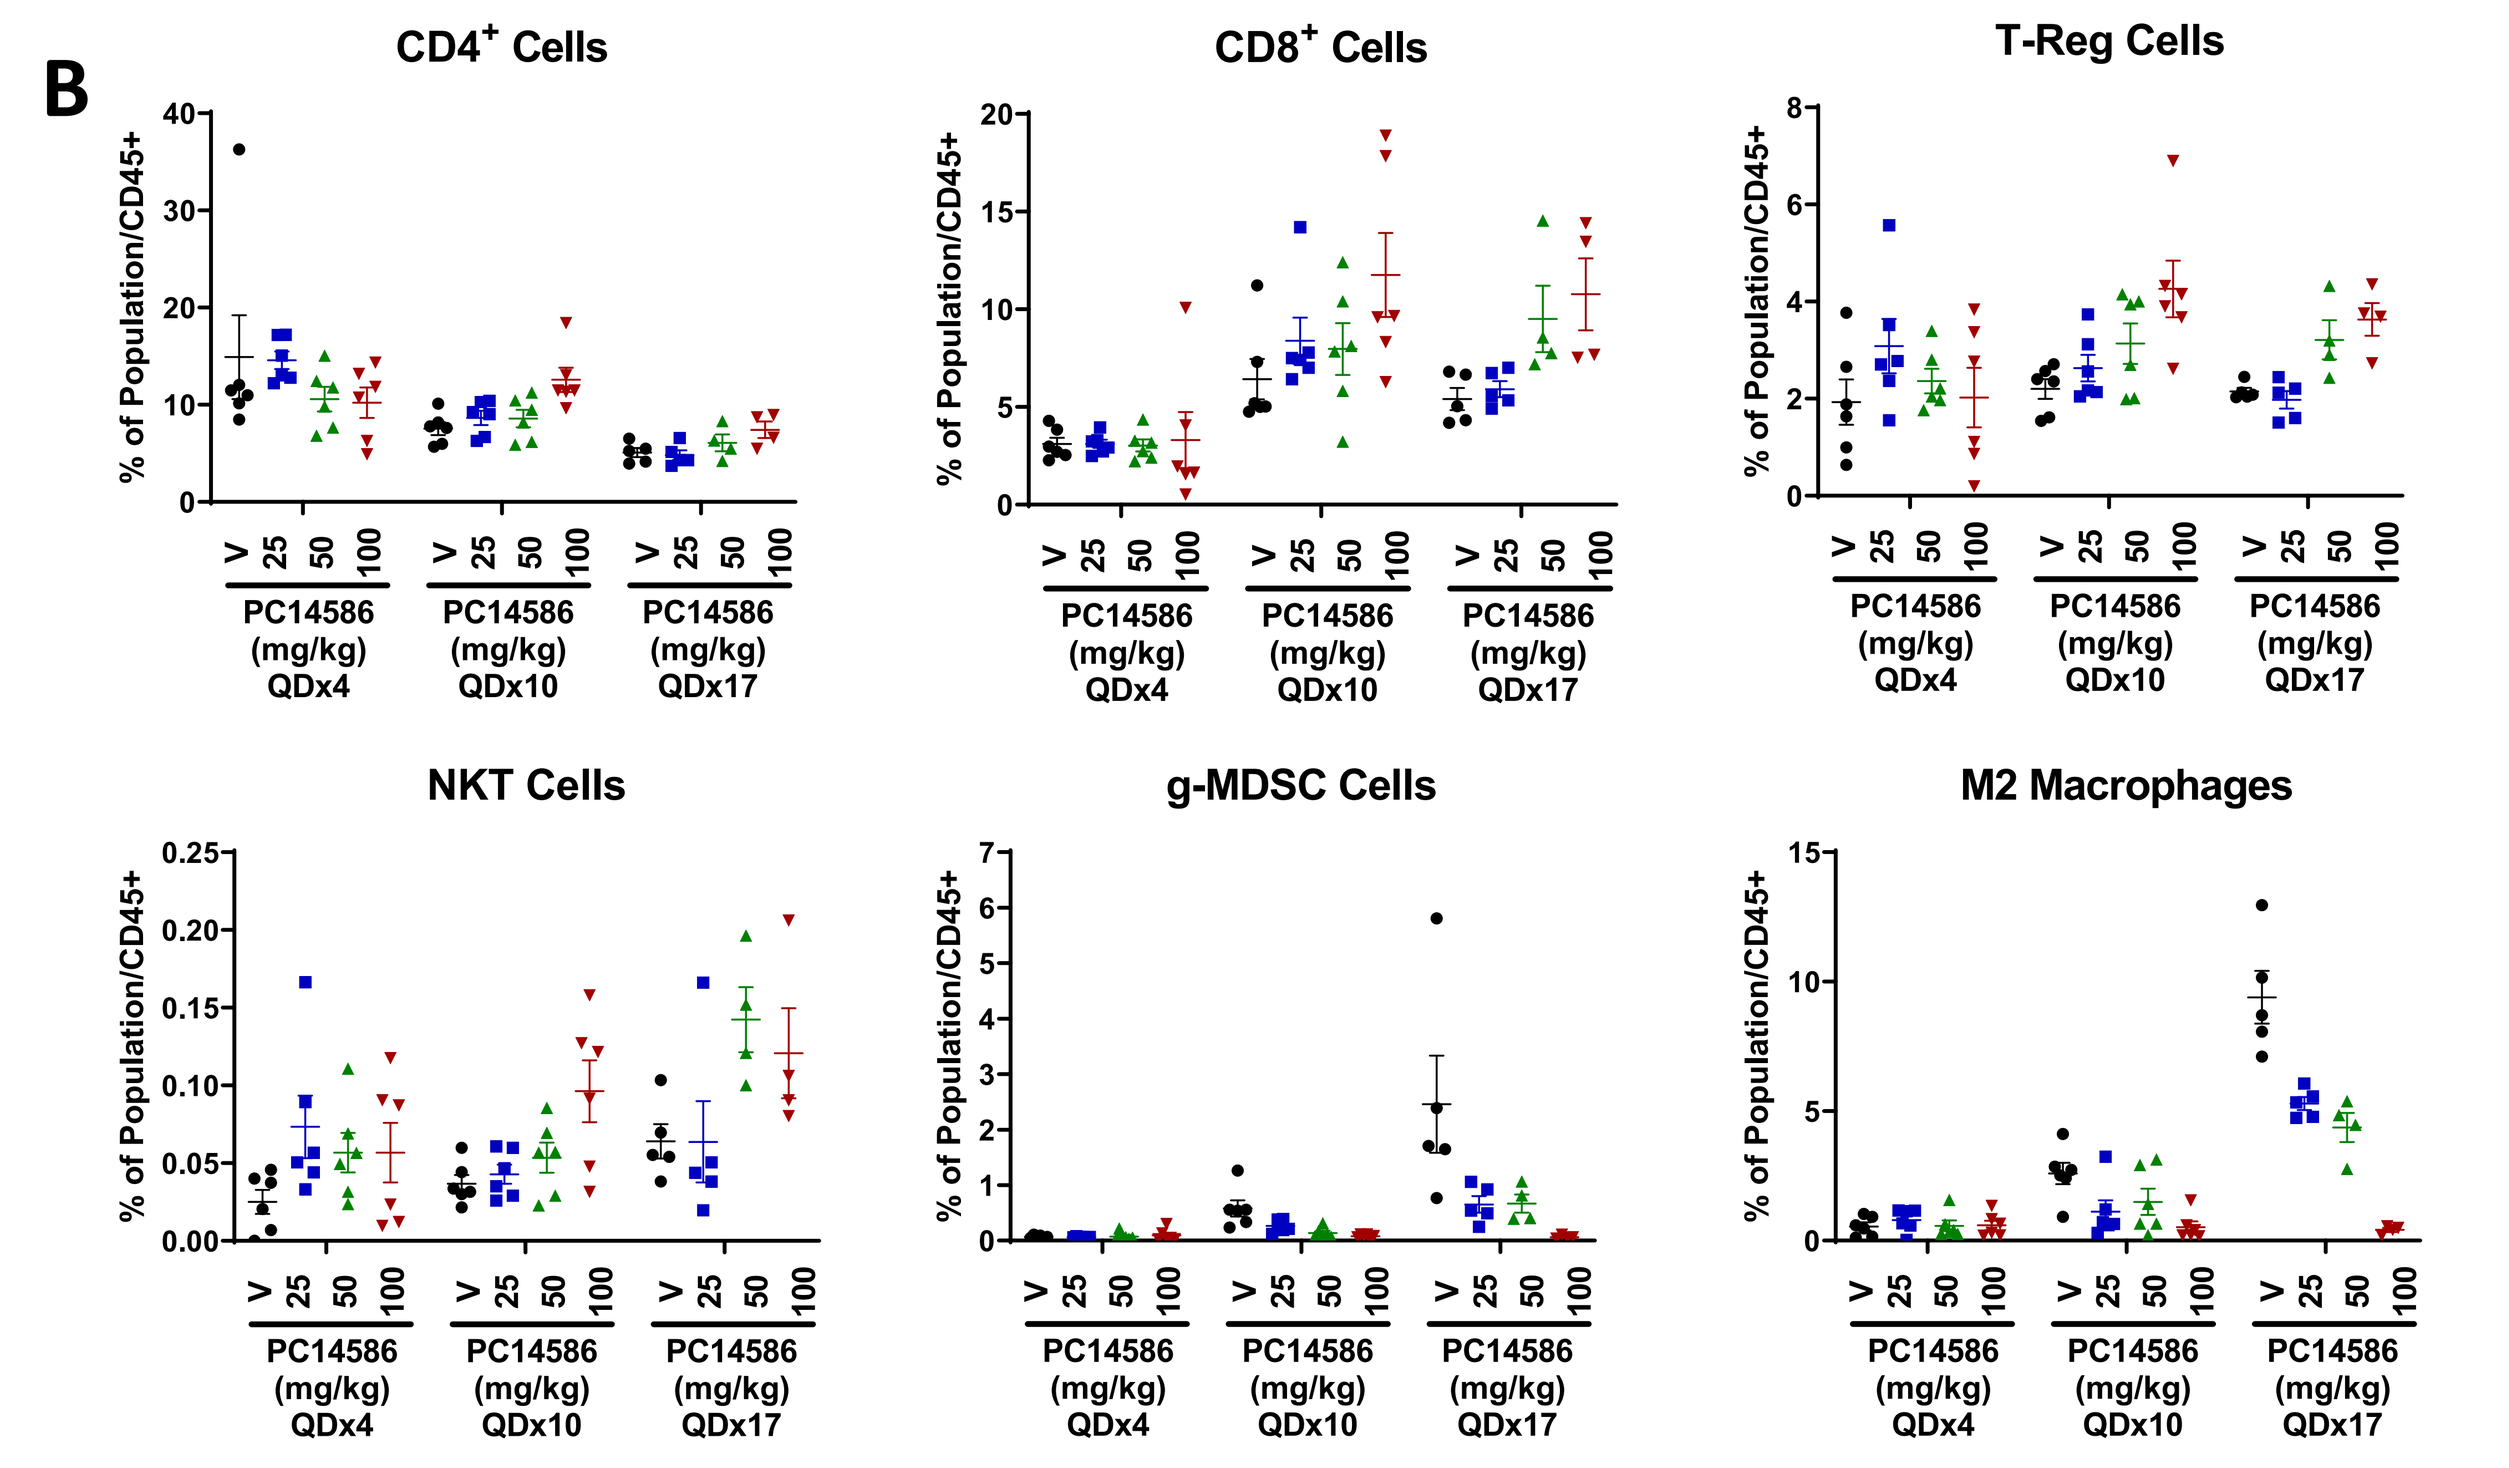
B
CD4+ Cells
CD8+ Cells
T-Reg Cells
NKT Cells
g-MDSC Cells
M2 Macrophages
% of Population/CD45+
PC14586 (mg/kg) QDx4
PC14586 (mg/kg) QDx10
PC14586 (mg/kg) QDx17

B. Six days post inoculation with MT373 cells, mice were administered PC14586 orally at 25, 50 or 100 mg/kg QD for 4, 10 or 17 doses and tumors harvested 24 h post dose for analysis of tumor infiltrating lymphocytes by flow cytometry. Reactivation of p53 lead to increases in $CD4^+$, $CD8^+$, T-Regulatory T-cells and NKT cells and decreases in g-MDSC and M2 macrophages that were dose responsive and time dependent.

Increase in T-Cell Signatures and Biomarkers of Checkpoint Activity Correlate with Synergistic Combination Effect

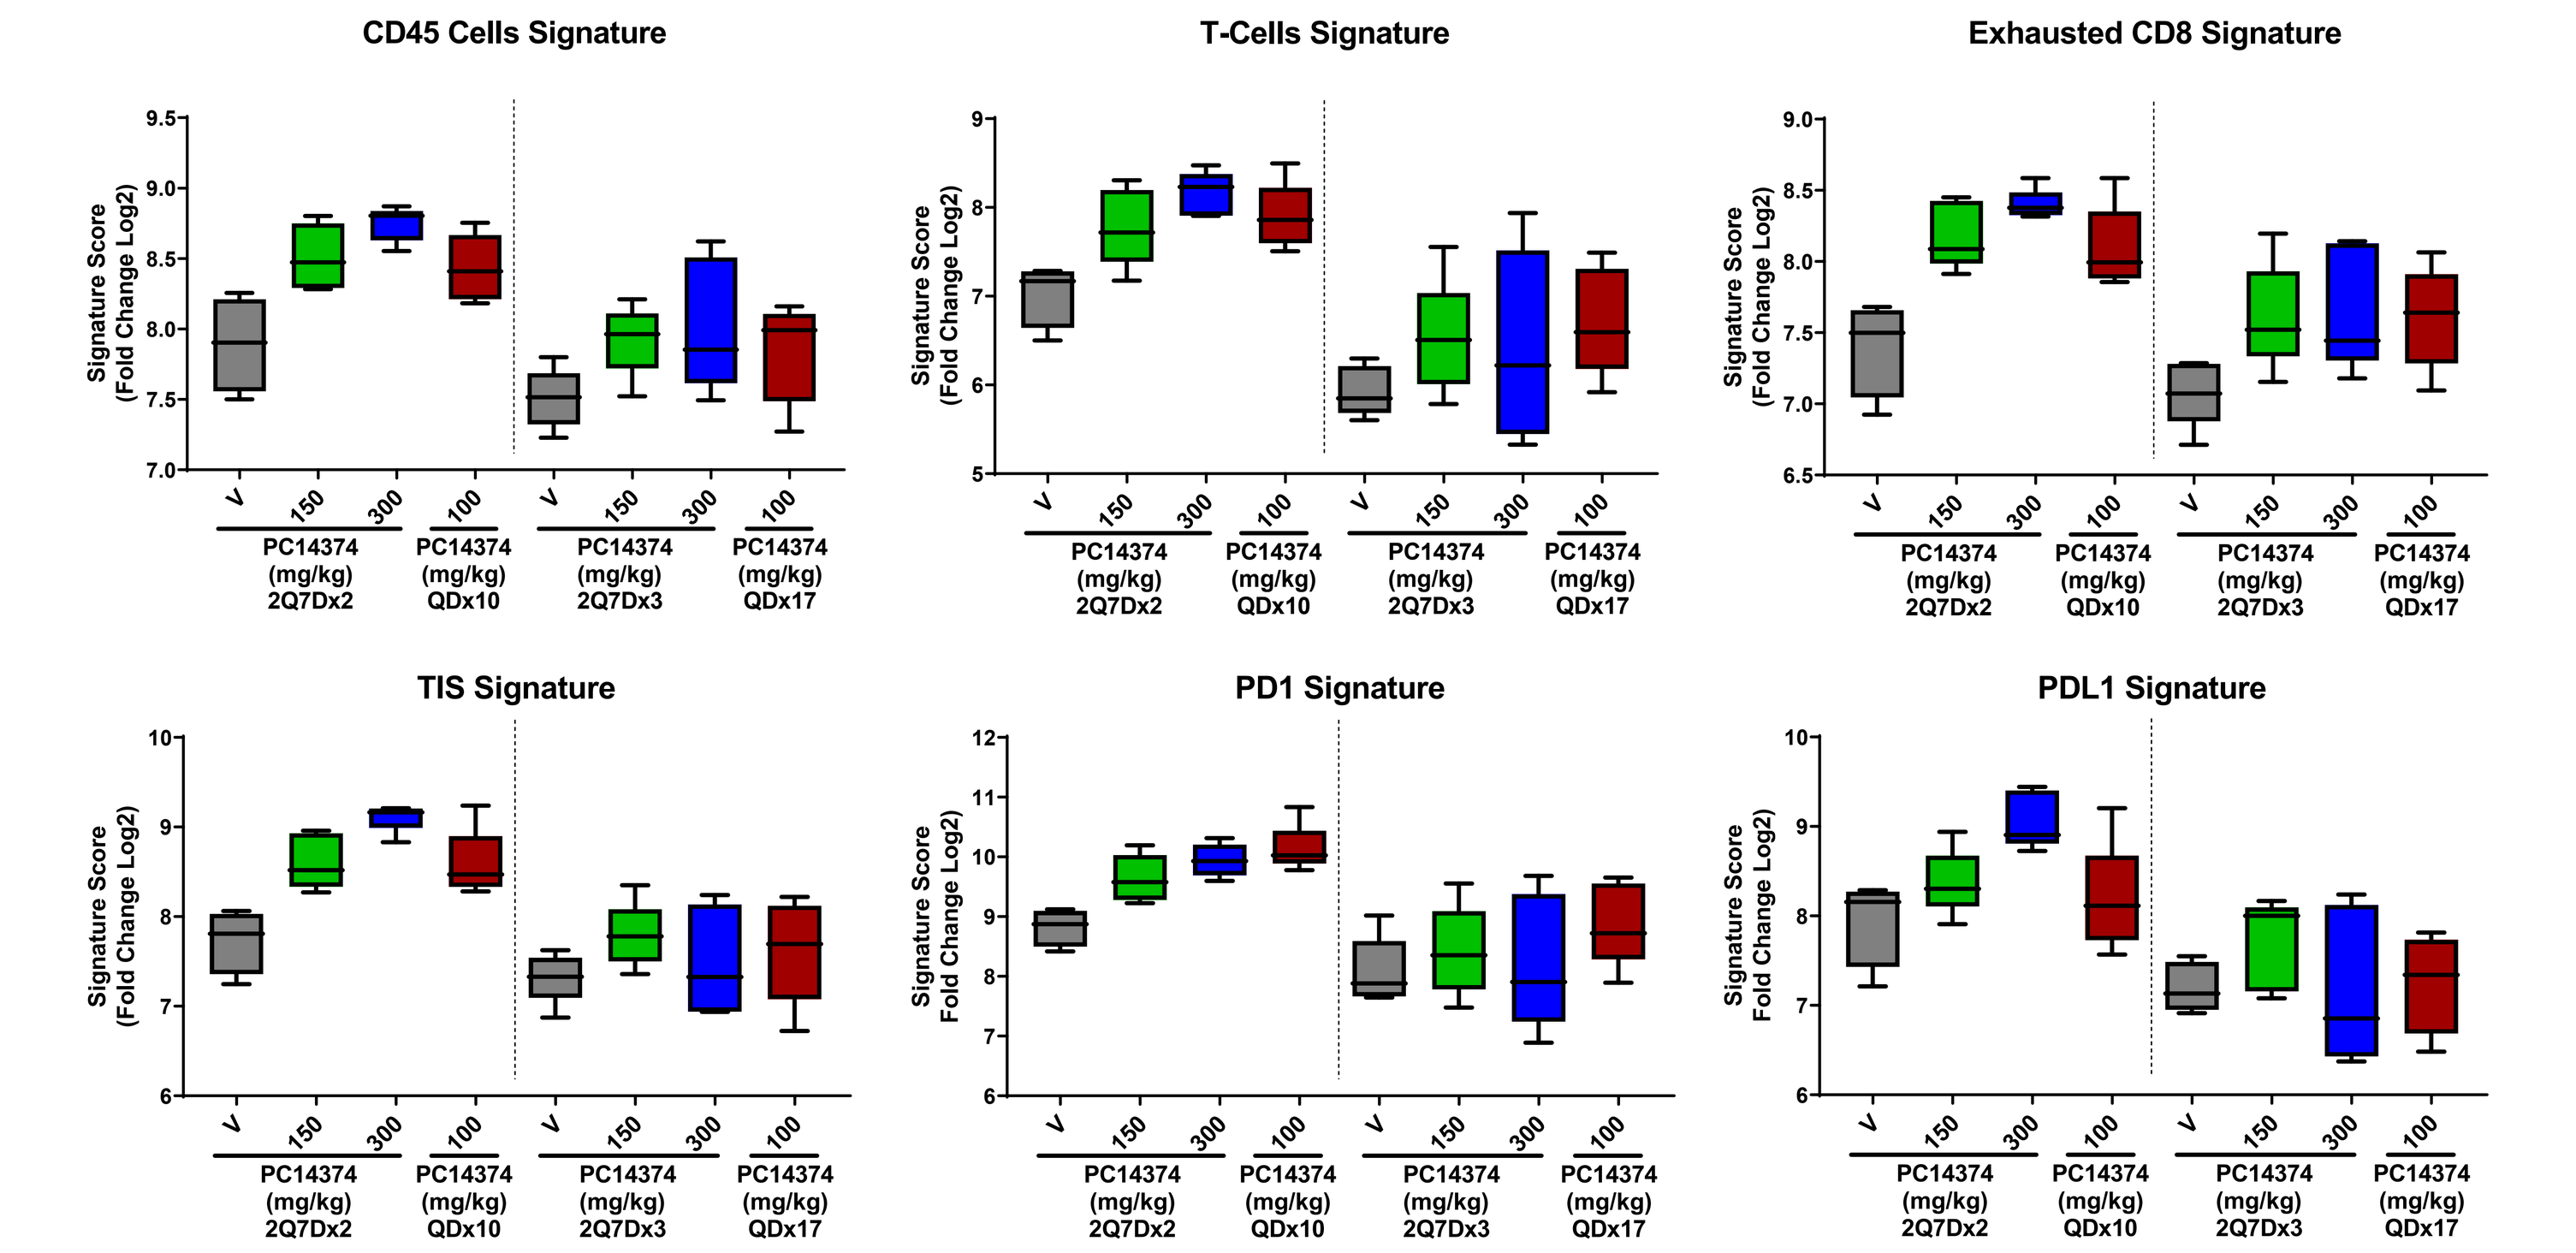
CD45 Cells Signature
T-Cells Signature
Exhausted CD8 Signature
TIS Signature
PD1 Signature
PDL1 Signature
Signature Score (Fold Change Log2)
PC14374 (mg/kg) 2Q7Dx2
PC14374 (mg/kg) QDx10
PC14374 (mg/kg) 2Q7Dx3
PC14374 (mg/kg) QDx17

FIGURE 5:
Six days post inoculation with MT373 cells, mice were administered PC14374 orally at 150 or 300 mg/kg 2Q7Dx2 or 2Q7Dx3 and 100 mg/kg QDx10 or QDx17; a similar dosing regimen as used in the flow cytometry analysis. Tumor samples were analyzed on the nanoString IO 360 panel for gene expression. Reactivation of p53 led to increases in CD45, T-cell and exhausted CD8 signatures that is consistent with the increase in similar populations as observed with flow cytometry and correlates with an immune response with p53 reactivation. Increases in Tumor Inflammation Signature (TIS), PD-1 signature, and PDL-1 signature with p53 reactivation are consistent with the synergistic effect seen with anti-PD-1 response in the combination efficacy studies (Figure 2).

$CD8^+$ Cell Depletion Decreases Anti-Tumor Effect with p53 Reactivation and Anti-PD-1 Combination

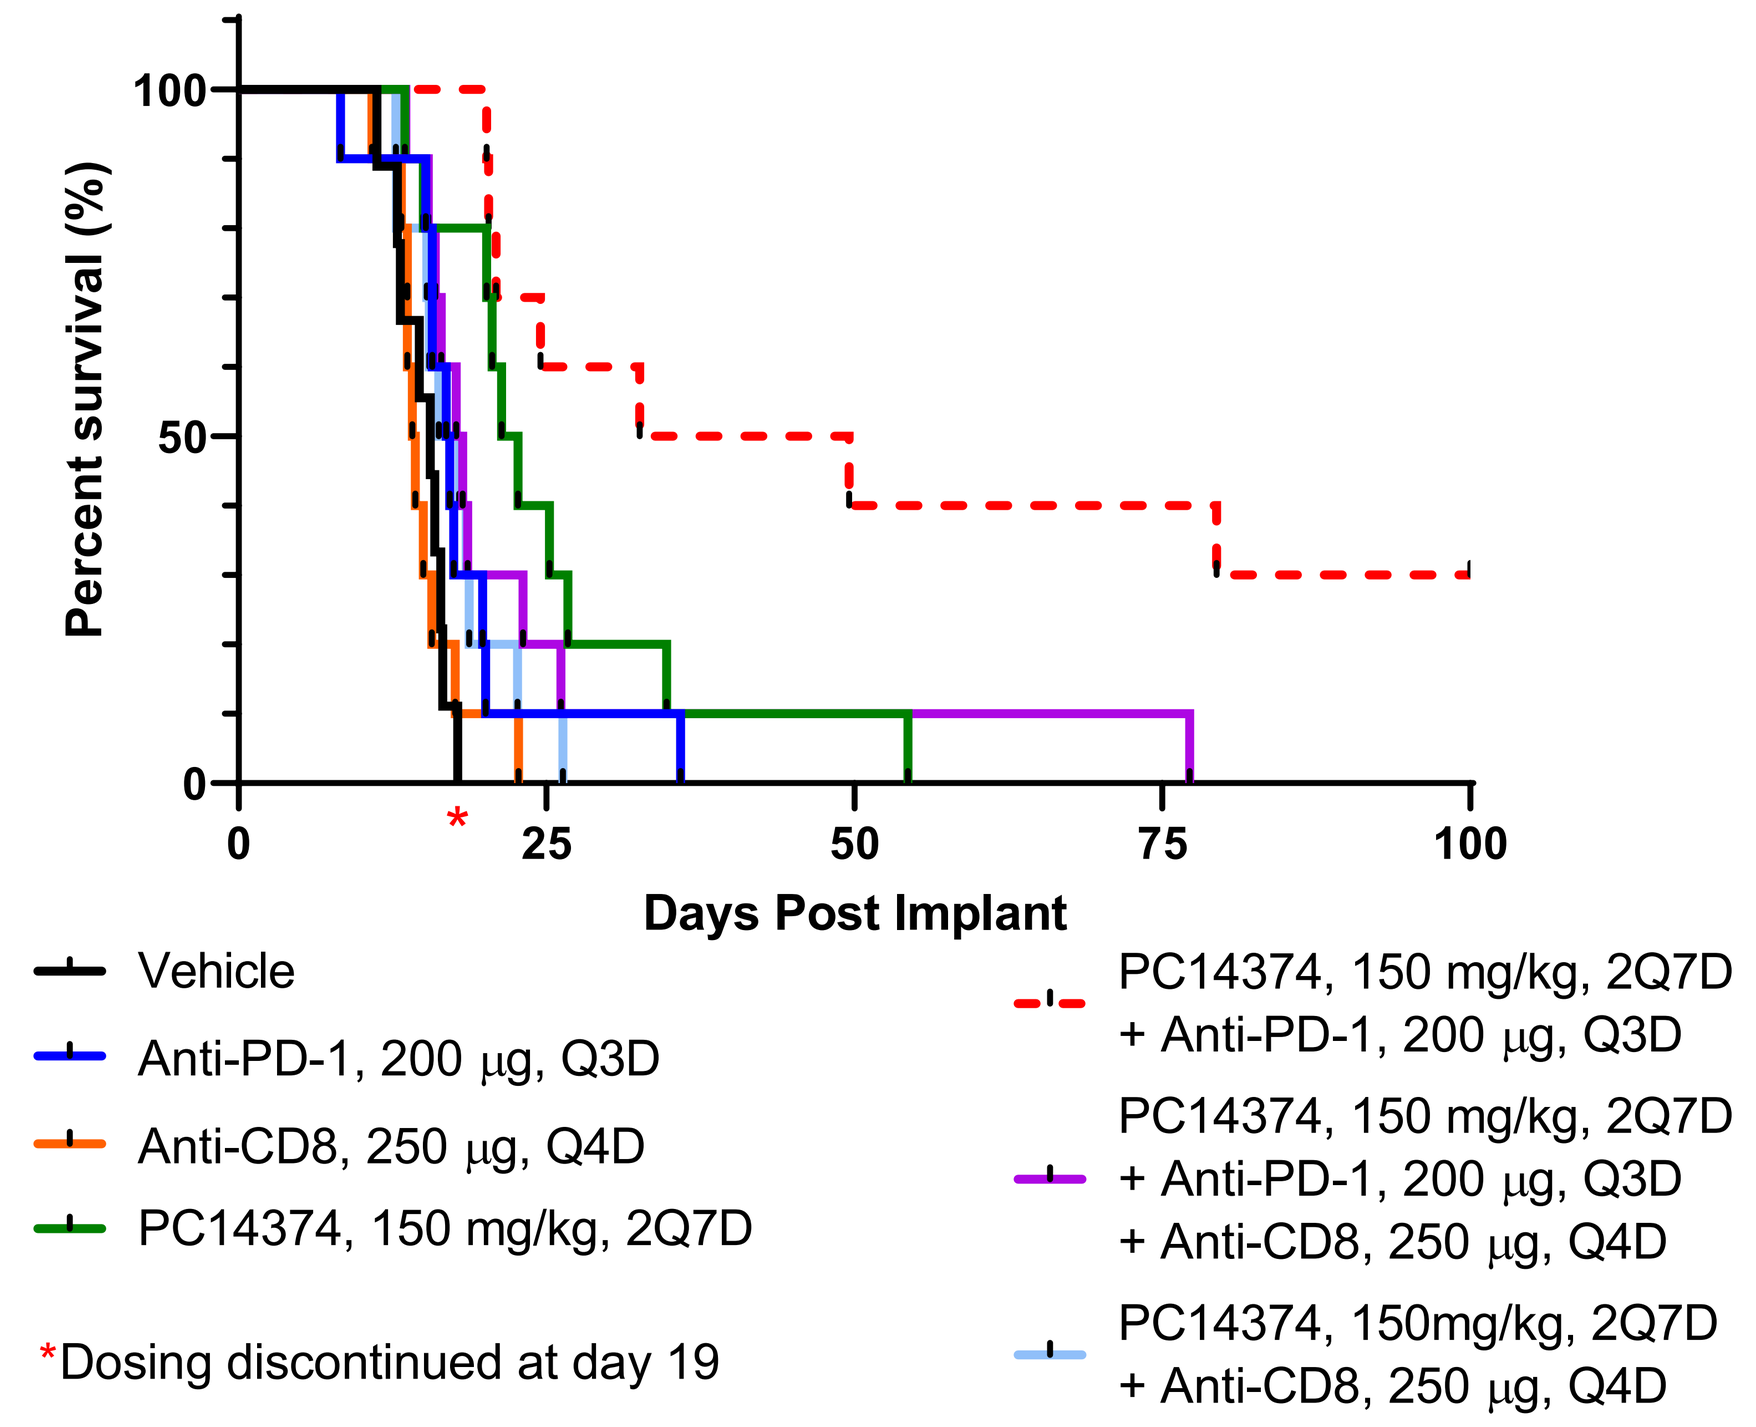
Percent survival (%)
Days Post Implant
Vehicle
Anti-PD-1, 200 µg, Q3D
Anti-CD8, 250 µg, Q4D
PC14374, 150 mg/kg, 2Q7D
*Dosing discontinued at day 19
PC14374, 150 mg/kg, 2Q7D + Anti-PD-1, 200 µg, Q3D
PC14374, 150 mg/kg, 2Q7D + Anti-PD-1, 200 µg, Q3D + Anti-CD8, 250 µg, Q4D
PC14374, 150mg/kg, 2Q7D + Anti-CD8, 250 µg, Q4D

FIGURE 6:
Six days post inoculation with MT373 cells, mice were administered a sub-efficacious dose of PC14374 orally, anti-PD-1 IP (BioXCell Clone RMPI-14) or anti-CD8 IP (BioXCell Clone 2.43), the combination of PC14374 with each antibody and the combination of the three. Mice in groups administered anti-CD8 were pre-treated 3 days prior to study start. The combination of PC14374 and anti-PD-1 results in a robust anti-tumor response that is lost with the addition of anti-CD8. Likewise, a similar reduction in anti-tumor response is measured when PC14374 is administered with anti-CD8 compared to without. Taken together the data suggests p53 reactivation leads to an increase in $CD8^+$ T-cells resulting in enhanced single agent activity of PC14374 and synergy in combination with anti-PD-1.

Summary of Results

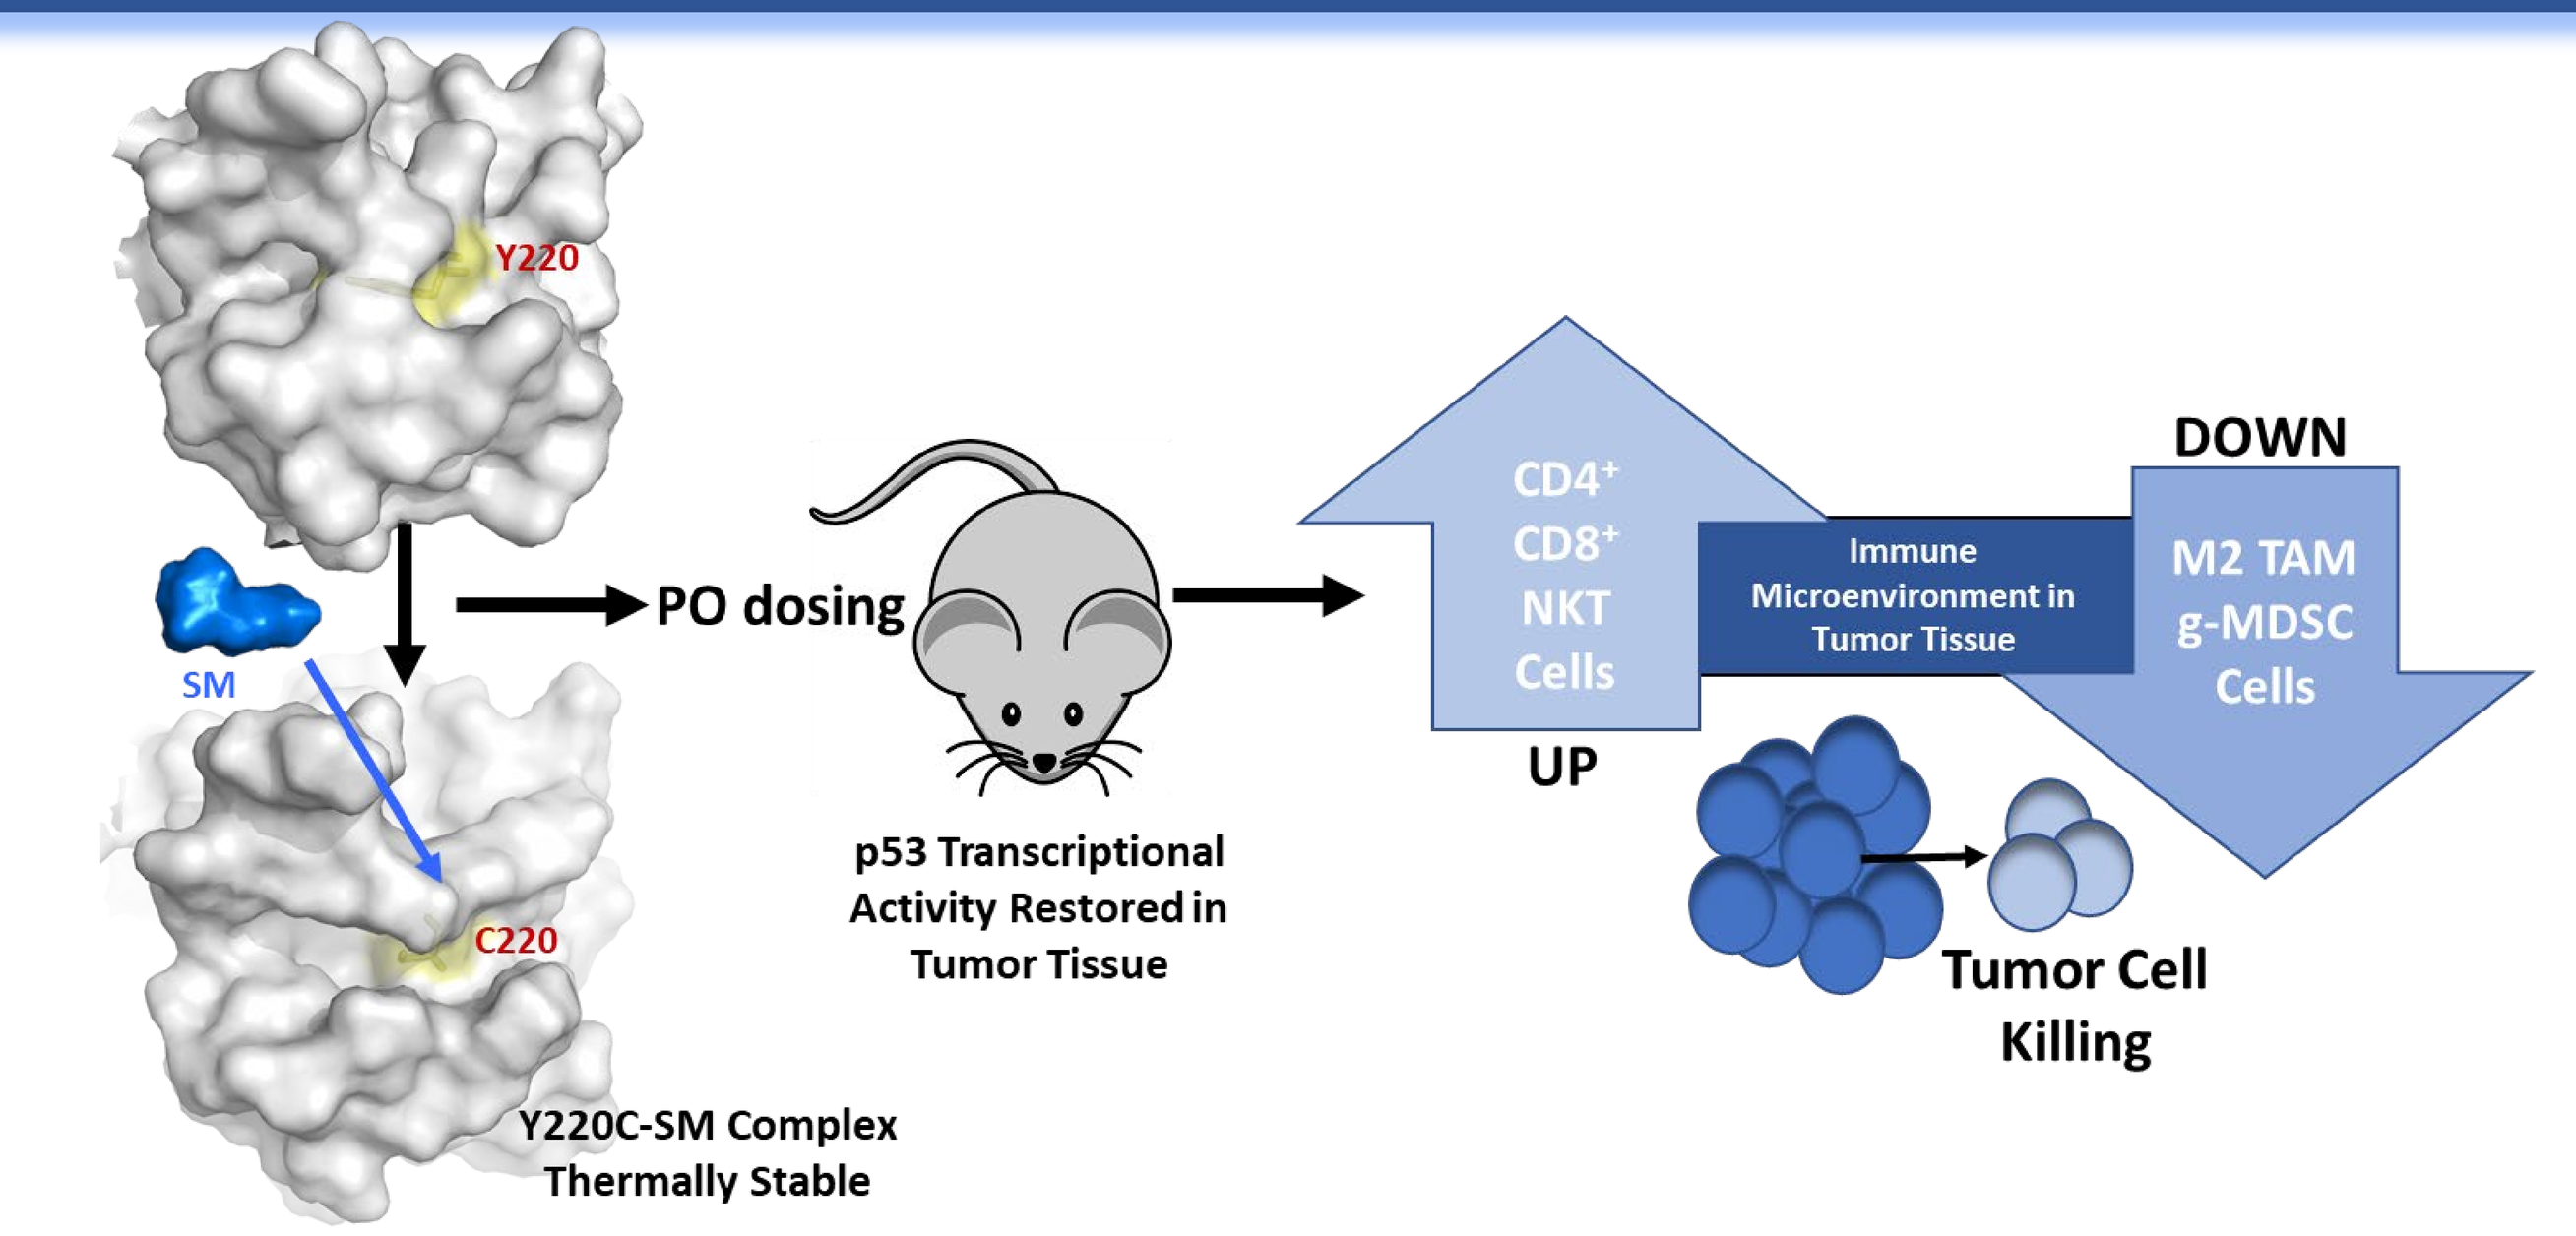
Y220
SM
C220
Y220C-SM Complex Thermally Stable
PO dosing
p53 Transcriptional Activity Restored in Tumor Tissue
CD4+
CD8+
NKT
Cells
UP
Immune Microenvironment in Tumor Tissue
DOWN
M2 TAM
g-MDSC
Cells
Tumor Cell Killing

Oral administration of p53 reactivators to mice, stabilizes Y220C p53 mutant protein in a WT conformation allowing the restoration of the p53 transcriptional activity specifically in the tumor tissue. In vivo studies have shown that p53 reactivation in the tumor leads to changes in intra-tumoral TIL cell populations, specifically increases in $CD4^+$, $CD8^+$, and NKT cells with decreases in M2 macrophages and g-MDSC cells. Administration of these p53 reactivators in combination with anti-PD-1 checkpoint therapy results in a synergistic anti-tumor effect. Depletion of $CD8^+$ T cells from mice results in a loss of this combination efficacy. Taken together, these data indicate that intra-tumoral p53 reactivation results in changes to the immune cell microenvironment that lead to highly synergistic efficacy when the reactivators are combined with checkpoint therapies.

Contact:

Anna Puzio-Kuter, PhD
PMV Pharmaceuticals
apuziokuter@pmvpharma.com
https://www.pmvpharma.com

Acknowledgments:

PMV would like to thank Guillermina Lozano's Lab for aging of HUPKI-Y220C mice and collecting tumors and cell line generation.

© Poster Template by Genigraphics · 1.800.790.4001 · www.genigraphics.com